Enhanced tumor control with a combination of APG-157 and immune checkpoint inhibitors for head and neck cancer

Daniel Sanghoon Shin[1,6,7], Mysore S. Veena[1], Saroj Bsak[2], Begoña Comin-Anduix[4,6], Tien S. Dong[3], Luis Avila[5], Marilene Wang[2,4], Eri Srivatsan[2,6,7]

1. Department of Medicine, Division of Hematology-Oncology, VAGLAHS/David Geffen School of Medicine at University of California, Los Angeles (UCLA), California (CA) . 2. Department of Surgery, VAGLAHS and Department of Head and Neck Surgery, David Geffen School of Medicine at UCLA, CA 3. Division of Digestive Diseases, Department of Medicine, David Geffen School of Medicine at UCLA 4. Department of Surgery, David Geffen School of Medicine at UCLA, CA . 5. Aveta-Biomics, Boston, MA. 6. Jonsson Comprehensive Cancer Center, Los Angeles, CA. 7. Member of Molecular Biology Institute, UCLA, CA

Acknowledgments

1. UCLA Department Medicine, Start up Fund.
2. Aveta-Biomics

Abstract 2652

OBJECTIVES

1. To investigate the anti-tumor activity of APG-157 in combination with immune checkpoint inhibitors.
2. Characterize tumor microenvironment with the combination treatment with APG-157 and immune checkpoint inhibitors
3. Assess gut microbiome changes by APG-157 combined with immunotherapy.

BACKGROUND

- Natural botanical drugs containing flavonoids are under clinical investigation for the treatment of malignancies.
- A phase I clinical study for patients with locally advanced head and neck cancer with a new class of botanical drug APG-157 showed excellent tolerability, systemic absorption via sublingual administration, and enhanced immune cell infiltration into the tumor microenvironment and suppression of T cell inhibitory inflammatory cytokine production.
- We sought to evaluate the efficacy of tumor control for the combination of APG-157 and checkpoint blockade immunotherapy using a head and neck cancer mouse model

METHODS

- CCL23 and UM-SCC1 head and neck cancer cell lines were utilized to assess interferon and NF-KB pathway with APG-157/other derivatives of curcumin treatments in presence or absence of interferons.
- SCCVII (HPV-) and MEER (HPV+) head and neck cancer murine cell line models were utilized in *in vivo* tumor growth study. After optimization for route and cell number, we measured tumor growth after treating mice with following groups: i) control, ii) APG-157, iii) anti-PD-1, iv) anti-CTLA-4, v) APG-157 + anti-PD-1, vi) APG-157 + anti-CTLA-4, vii) anti-PD-1 and anti-CTLA-4 , viii) APG-157 + anti-PD-1 + anti-CTLA-4. Tumors were collected after 2 injections of immune checkpoint inhibitors and characterized for immunophenotype by multi-color flow cytometry, immunohistochemistry, and RNA-sequence analysis. Fecal microbiome was analyzed by 16S rRNA sequence for SCCVII model to assess the impact of APG-157 on the microbiome as we observed the shift of oral microbiome species in the human trial. (Figure 1 for *in vivo* study diagram.)

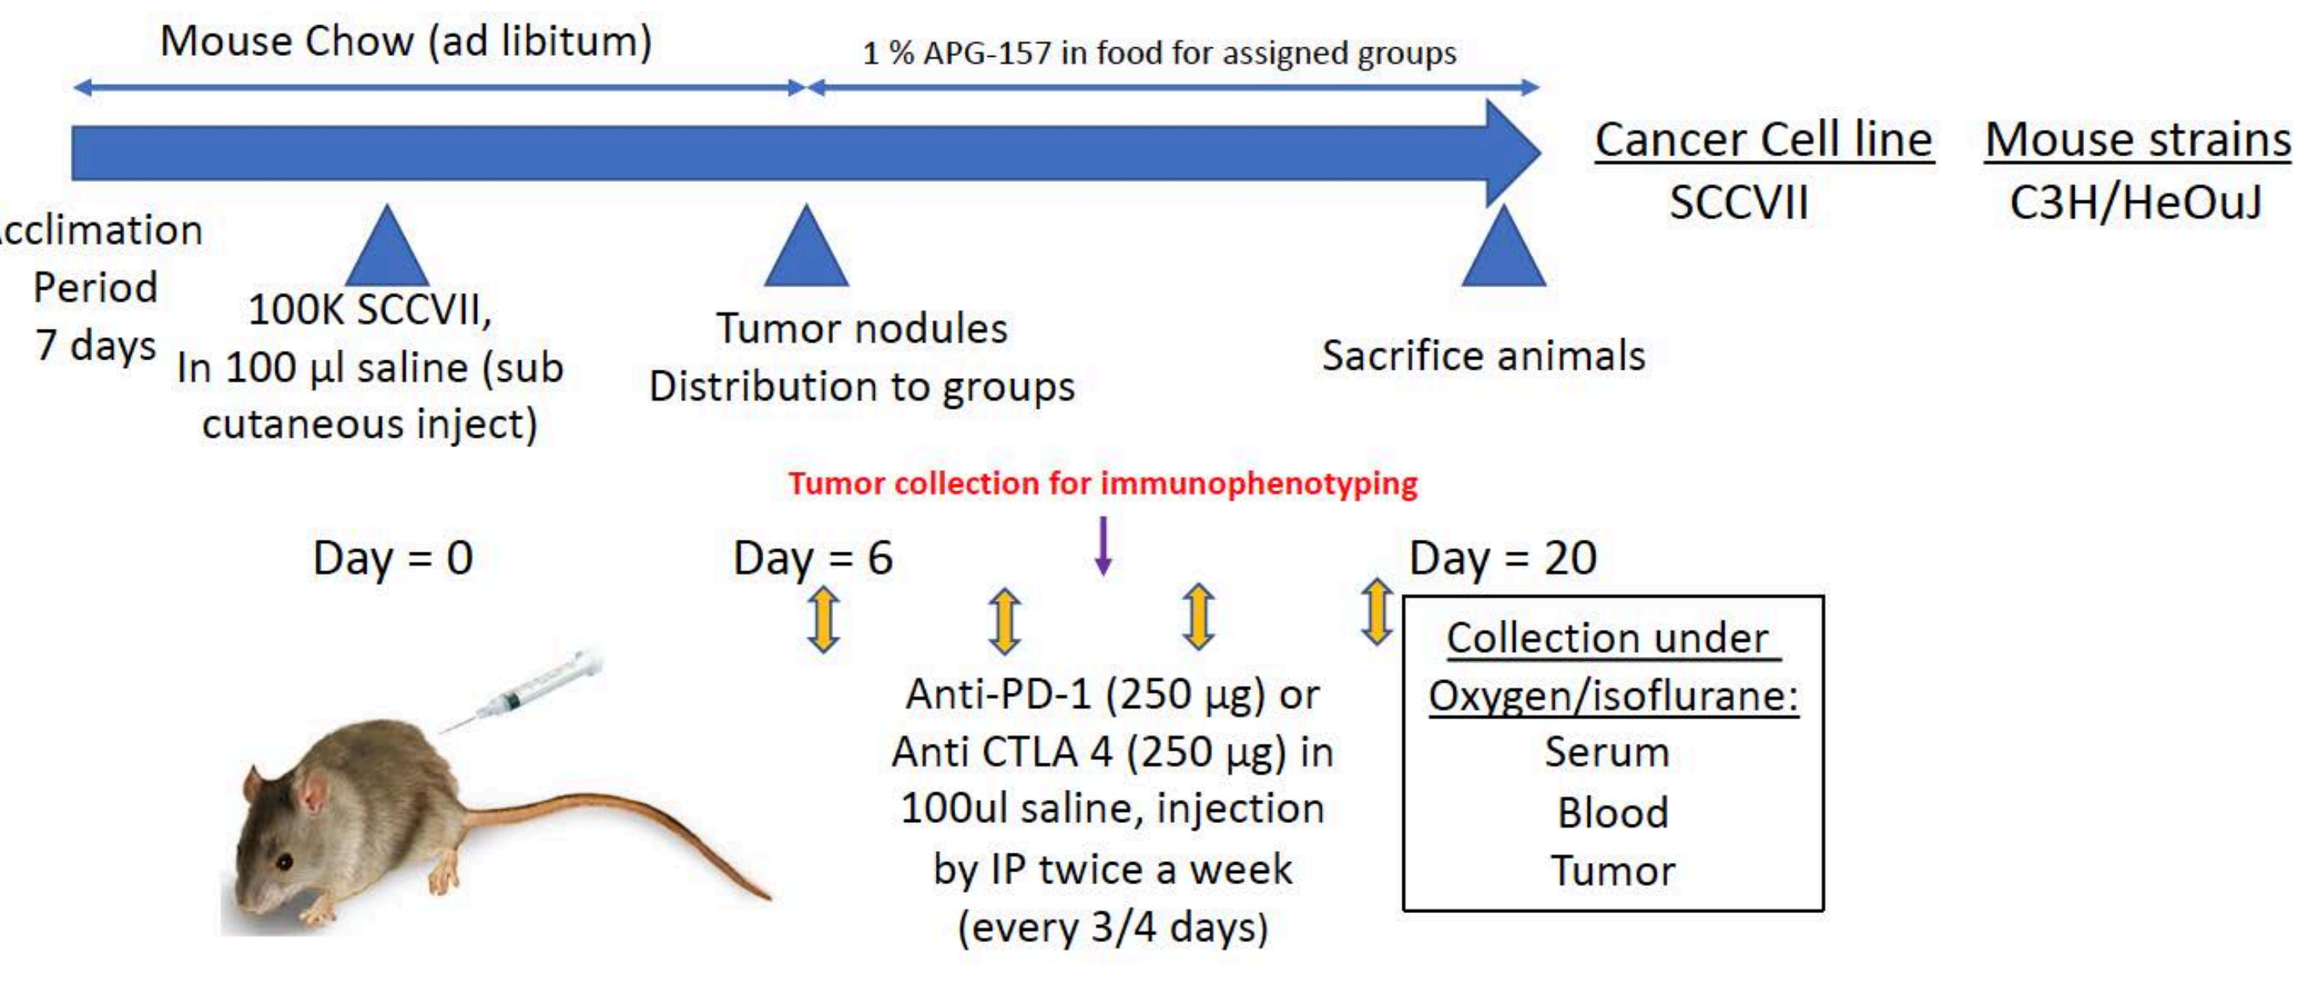
Mouse Chow (ad libitum)
1 % APG-157 in food for assigned groups
Cancer Cell line
SCCVII
Mouse strains
C3H/HeOuJ
Acclimation Period 7 days
100K SCCVII, In 100 µl saline (sub cutaneous inject)
Tumor nodules Distribution to groups
Sacrifice animals
Tumor collection for immunophenotyping
Day = 0
Day = 6
Day = 20
Anti-PD-1 (250 µg) or Anti CTLA 4 (250 µg) in 100ul saline, injection by IP twice a week (every 3/4 days)
Collection under Oxygen/isoflurane:
Serum
Blood
Tumor

Figure 1: Diagram of *in vivo* study using SCCVII murine head and neck cancer cell line model.

RESULTS

Figure 2: APG-157 and curcumin derivatives suppress the proliferation of human head and neck cancer cells

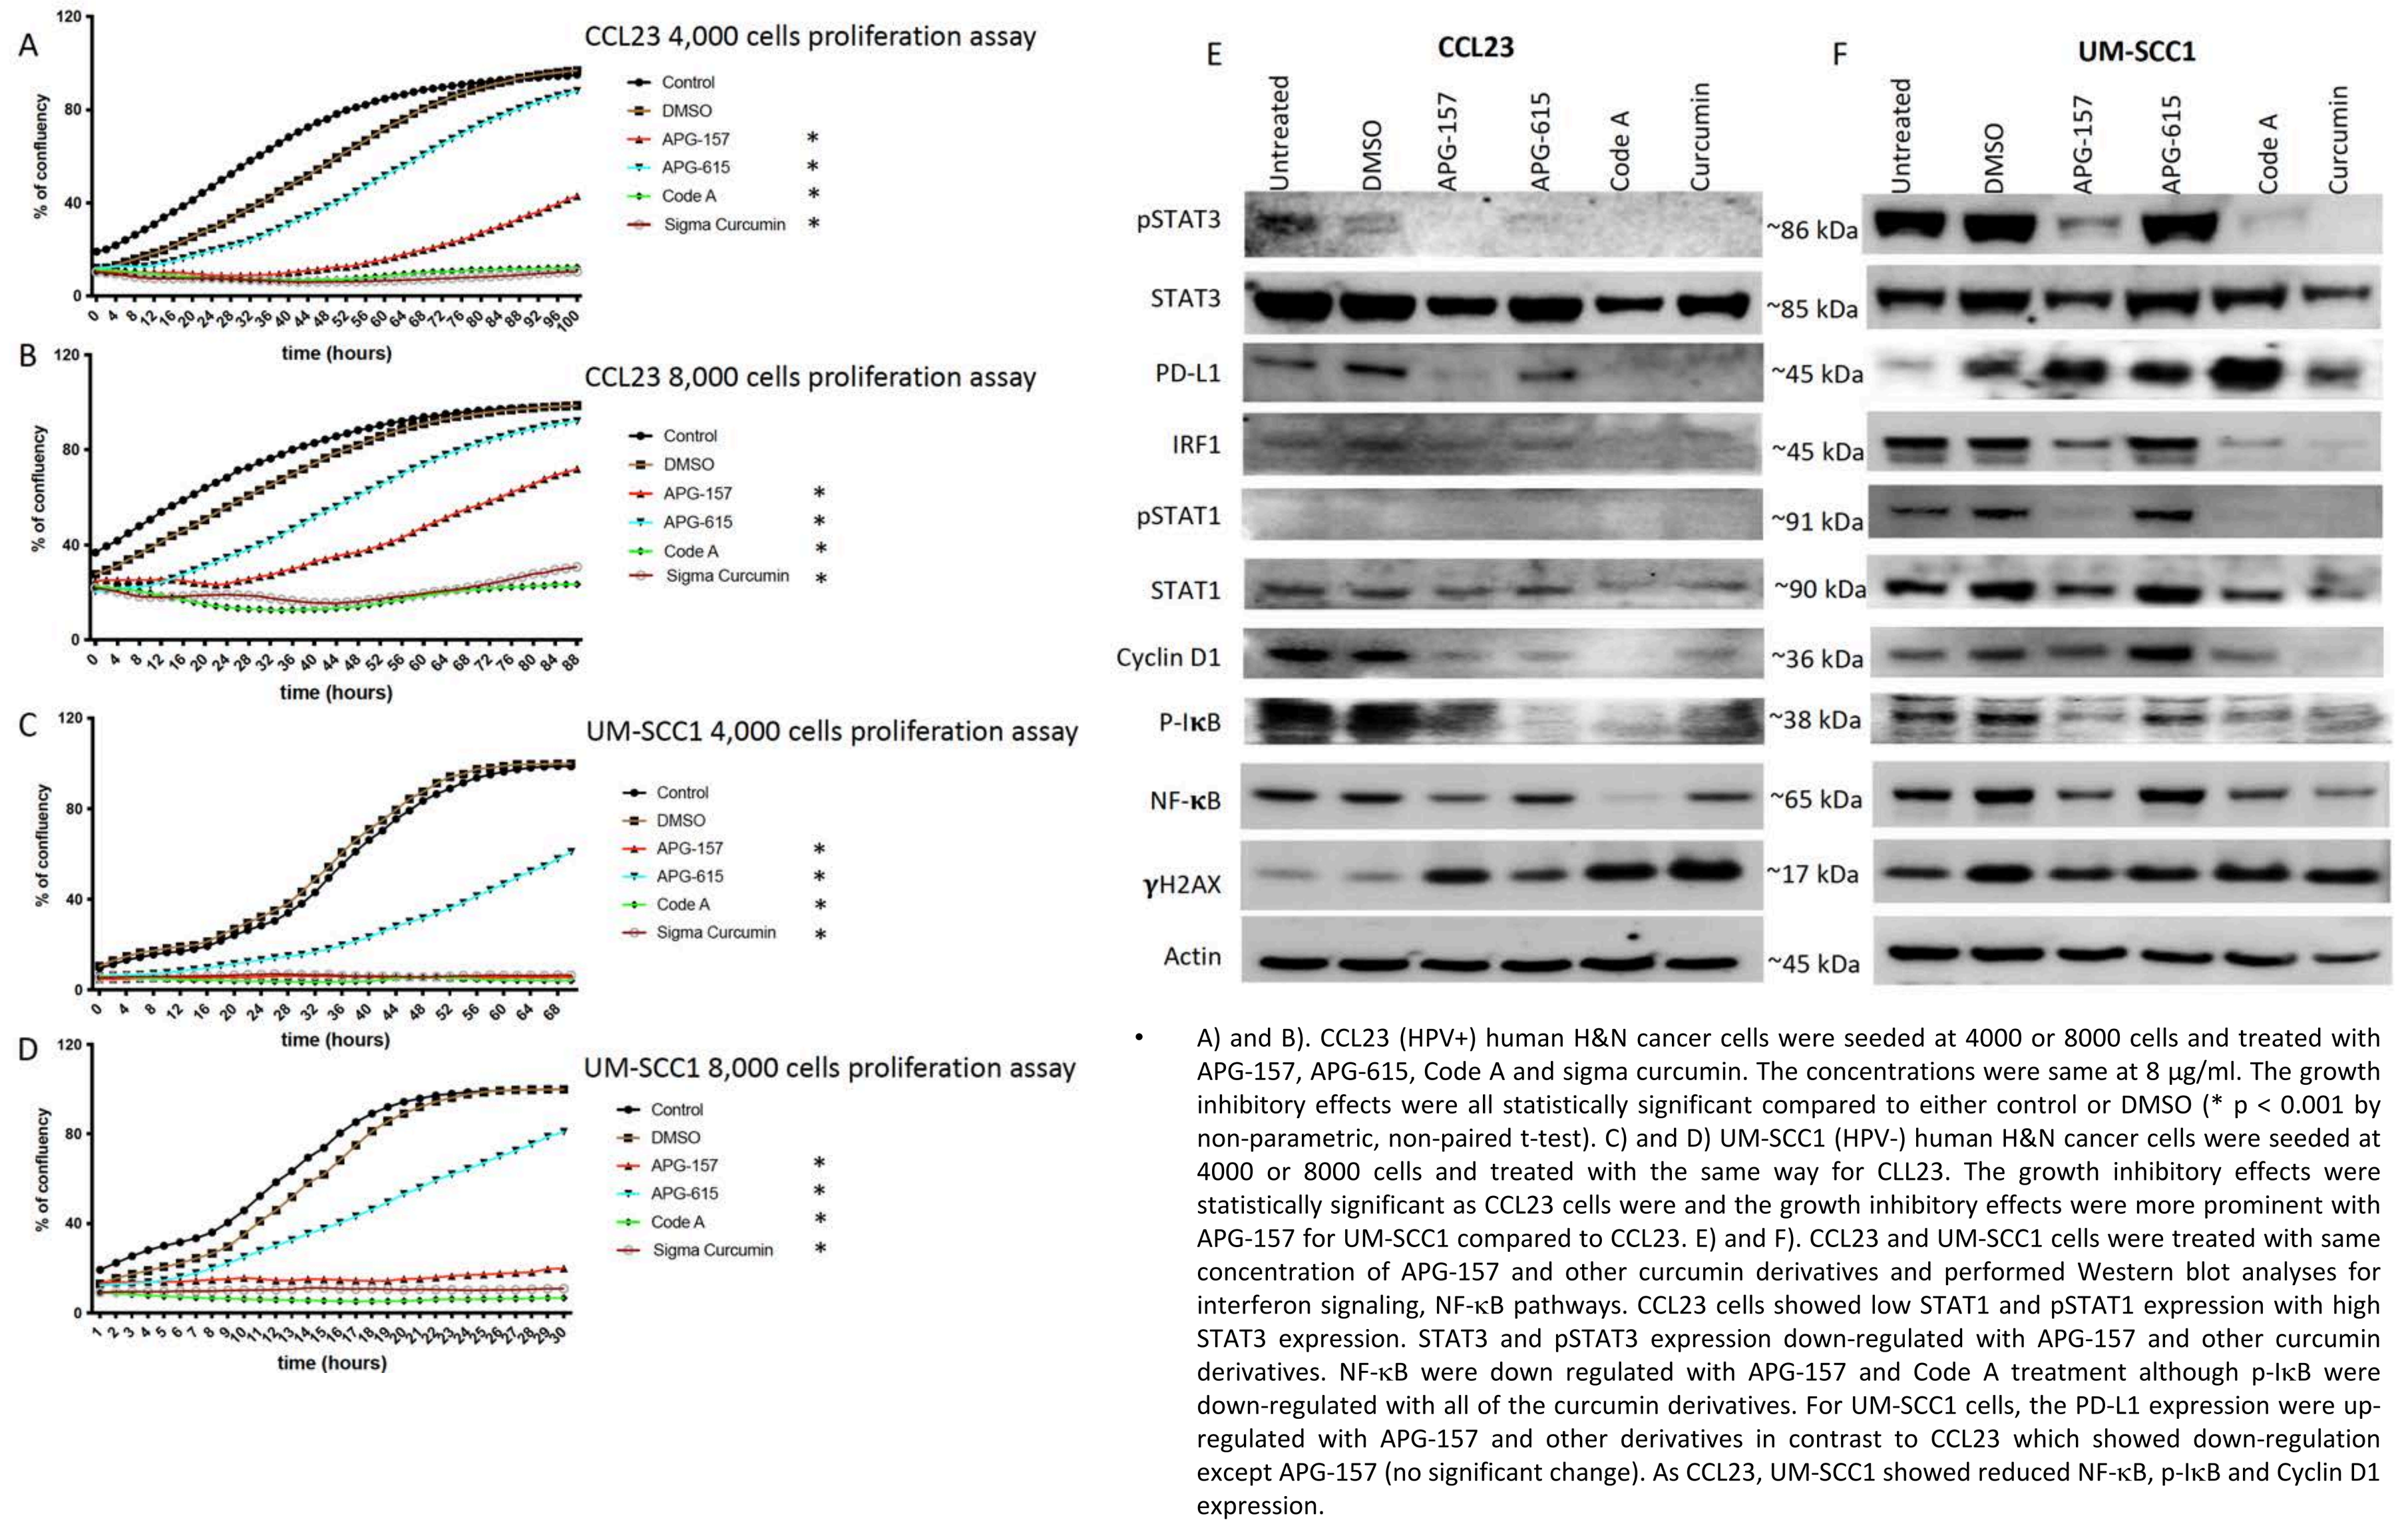
CCL23 4,000 cells proliferation assay
CCL23 8,000 cells proliferation assay
UM-SCC1 4,000 cells proliferation assay
UM-SCC1 8,000 cells proliferation assay
Control
DMSO
APG-157
APG-615
Code A
Sigma Curcumin
time (hours)
% of confluency
CCL23
UM-SCC1
Untreated
DMSO
APG-157
APG-615
Code A
Curcumin
pSTAT3
STAT3
PD-L1
IRF1
pSTAT1
STAT1
Cyclin D1
P-IκB
NF-κB
γH2AX
Actin

- A) and B). CCL23 (HPV+) human H&N cancer cells were seeded at 4000 or 8000 cells and treated with APG-157, APG-615, Code A and sigma curcumin. The concentrations were same at 8 µg/ml. The growth inhibitory effects were all statistically significant compared to either control or DMSO (* p < 0.001 by non-parametric, non-paired t-test). C) and D) UM-SCC1 (HPV-) human H&N cancer cells were seeded at 4000 or 8000 cells and treated with the same way for CLL23. The growth inhibitory effects were statistically significant as CCL23 cells were and the growth inhibitory effects were more prominent with APG-157 for UM-SCC1 compared to CCL23. E) and F). CCL23 and UM-SCC1 cells were treated with same concentration of APG-157 and other curcumin derivatives and performed Western blot analyses for interferon signaling, NF-κB pathways. CCL23 cells showed low STAT1 and pSTAT1 expression with high STAT3 expression. STAT3 and pSTAT3 expression down-regulated with APG-157 and other curcumin derivatives. NF-κB were down regulated with APG-157 and Code A treatment although p-IκB were down-regulated with all of the curcumin derivatives. For UM-SCC1 cells, the PD-L1 expression were up-regulated with APG-157 and other derivatives in contrast to CCL23 which showed down-regulation except APG-157 (no significant change). As CCL23, UM-SCC1 showed reduced NF-κB, p-IκB and Cyclin D1 expression.

Figure 3: Anti-tumor activity of APG-157 and combination treatment with ICI

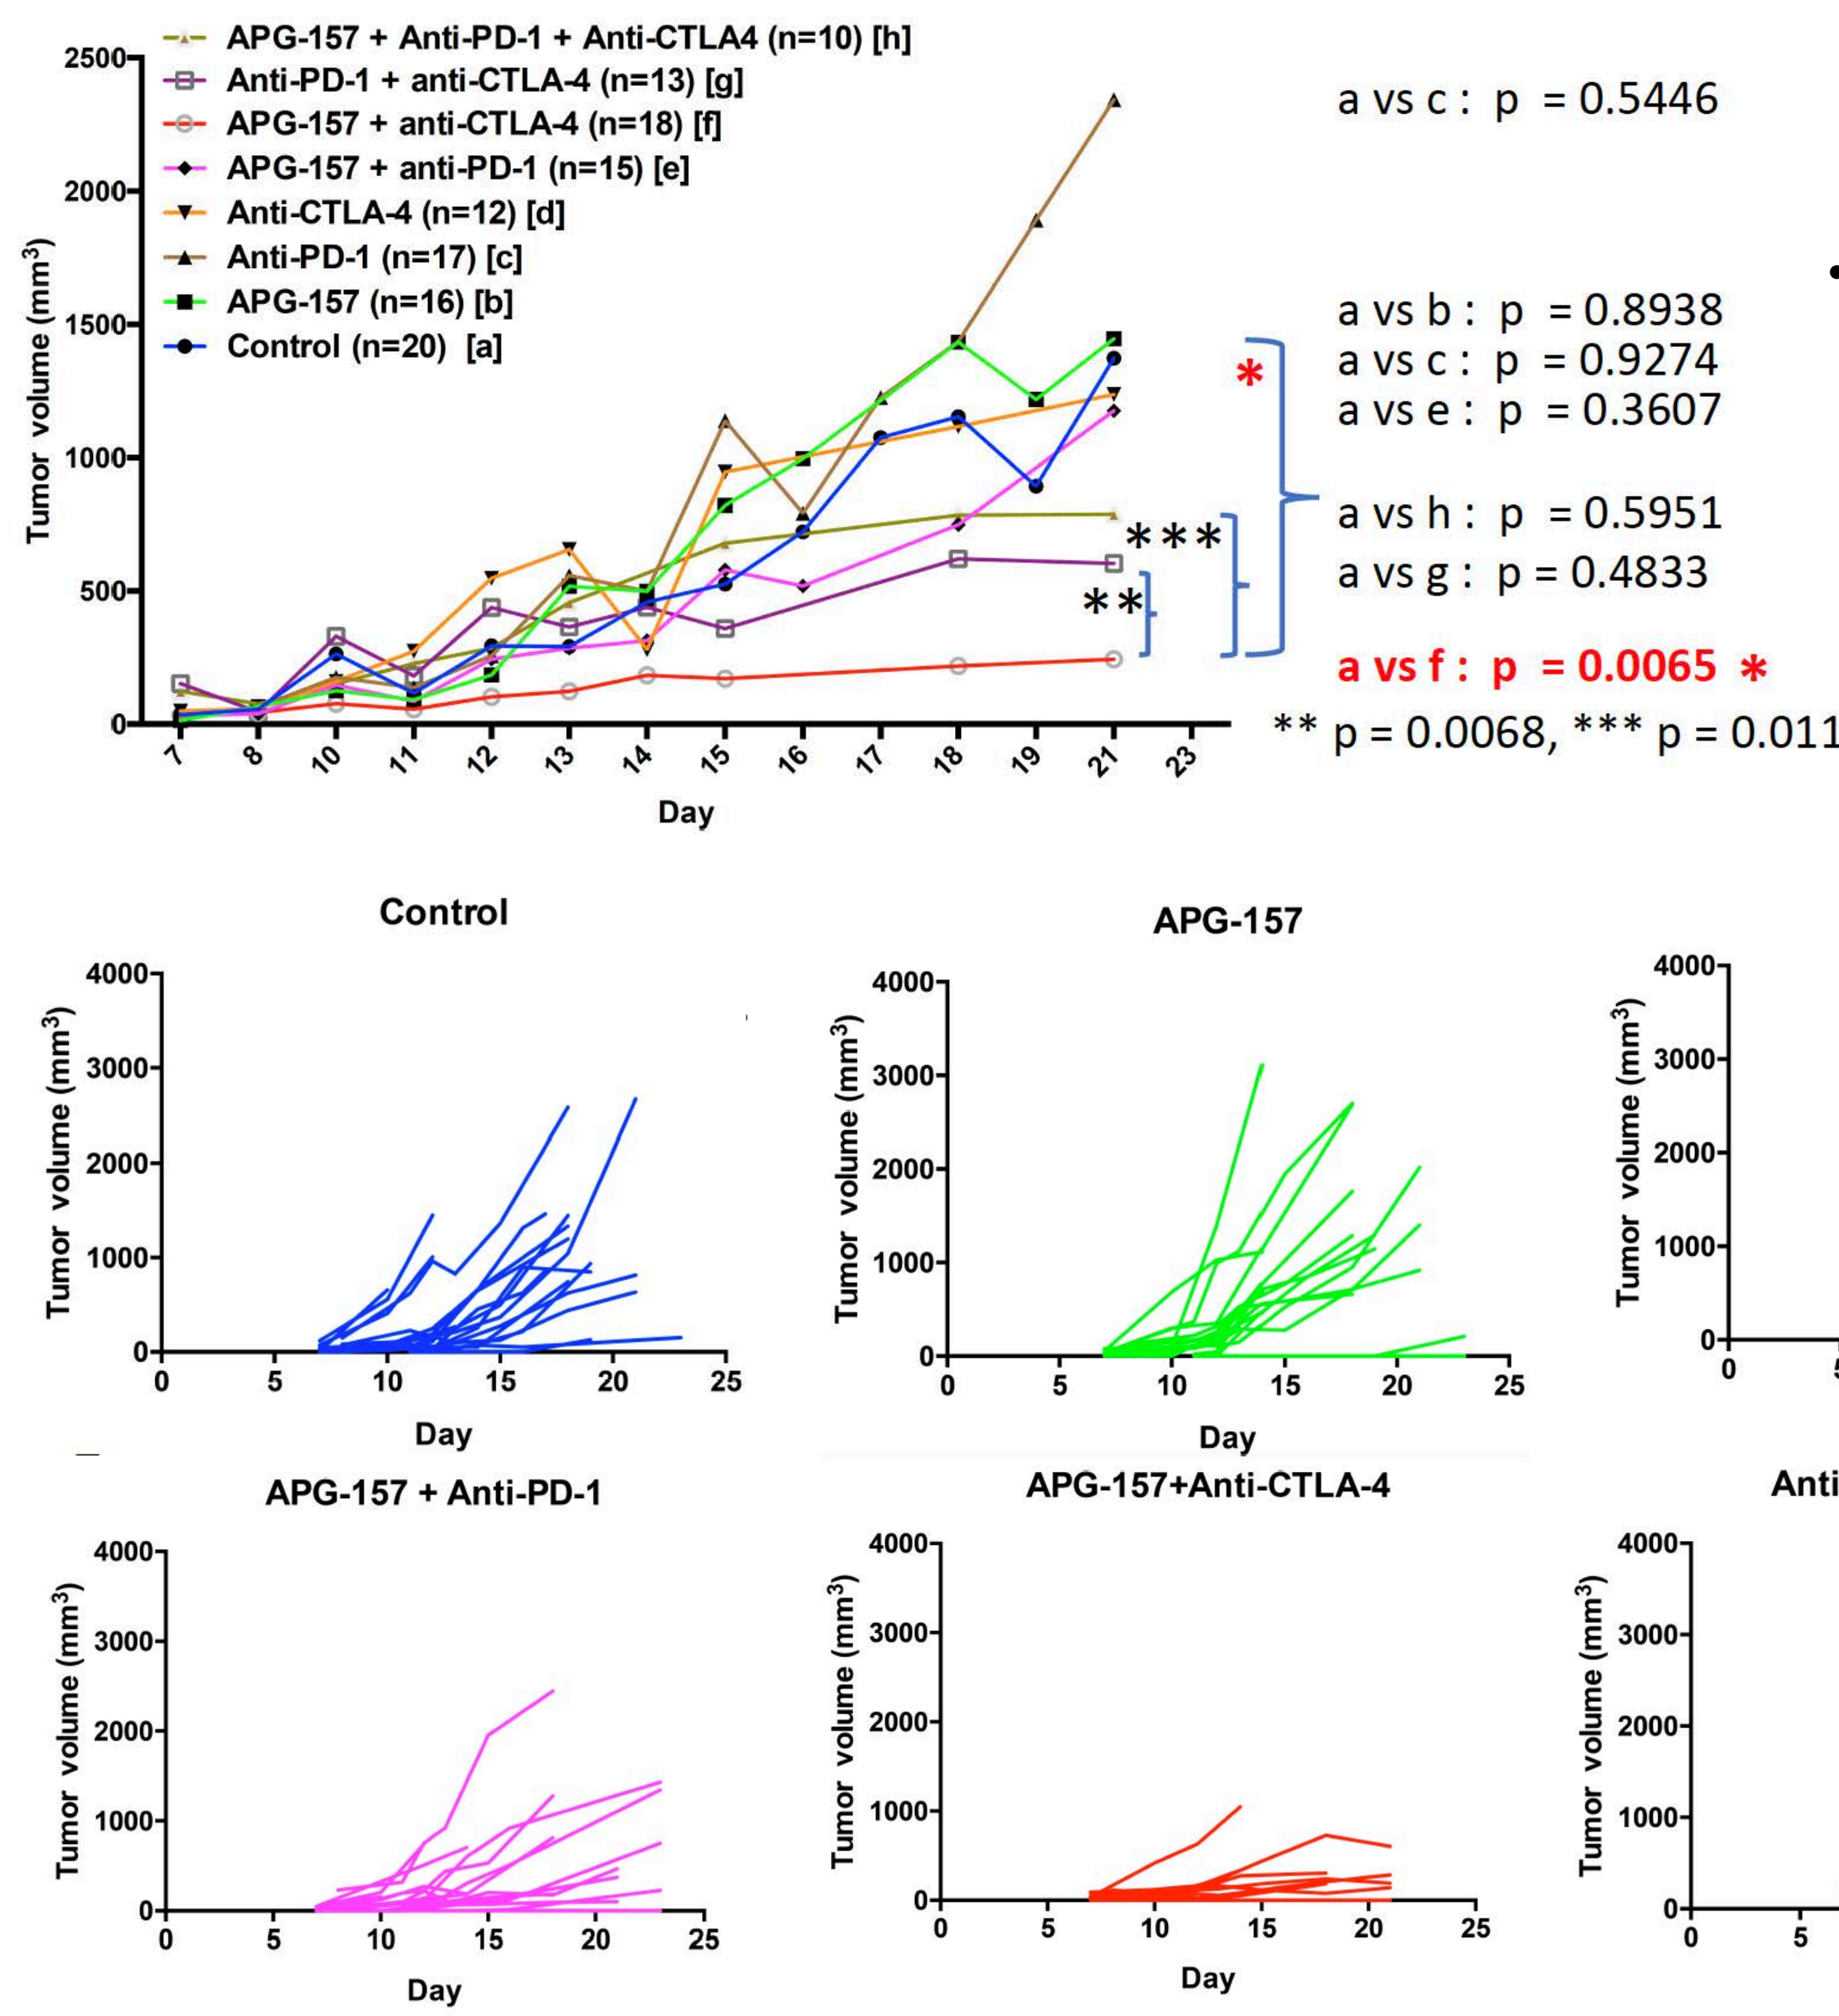
APG-157 + Anti-PD-1 + Anti-CTLA4 (n=10) [h]
Anti-PD-1 + anti-CTLA-4 (n=13) [g]
APG-157 + anti-CTLA-4 (n=18) [f]
APG-157 + anti-PD-1 (n=15) [e]
Anti-CTLA-4 (n=12) [d]
Anti-PD-1 (n=17) [c]
APG-157 (n=16) [b]
Control (n=20) [a]
Tumor volume (mm³)
Day
a vs c : p = 0.5446
a vs b : p = 0.8938
a vs c : p = 0.9274
a vs e : p = 0.3607
a vs h : p = 0.5951
a vs g : p = 0.4833
a vs f : p = 0.0065 *
** p = 0.0068, *** p = 0.0117
Control
APG-157
Anti-PD-1
Anti-CTLA-4
APG-157 + Anti-PD-1
APG-157+Anti-CTLA-4
Anti-PD-1+Anti-CTLA-4
APG-157+Anti-CTLA-4+Anti-PD-1

- The best tumor control group was APG-157 and anti-CTLA-4 antibody combination treatment compared to control (red-color statistics).
- The statistical analysis was performed by Prism graph pad program, non-paired, non-parametric two-sided, Mann-Whitney t-test. The difference between treatment groups, APG-157 + anti-CTLA-4 vs Anti-PD-1+ anti-CTLA-4 or APG-157 + anti-CTLA-4 vs APG-157 + anti-PD-1 + anti-CTLA-4 were significantly different as the statistics show (double and triple asterix).

Figure 4: Increased T cell infiltration into Tumor Microenvironment assessed by flow.

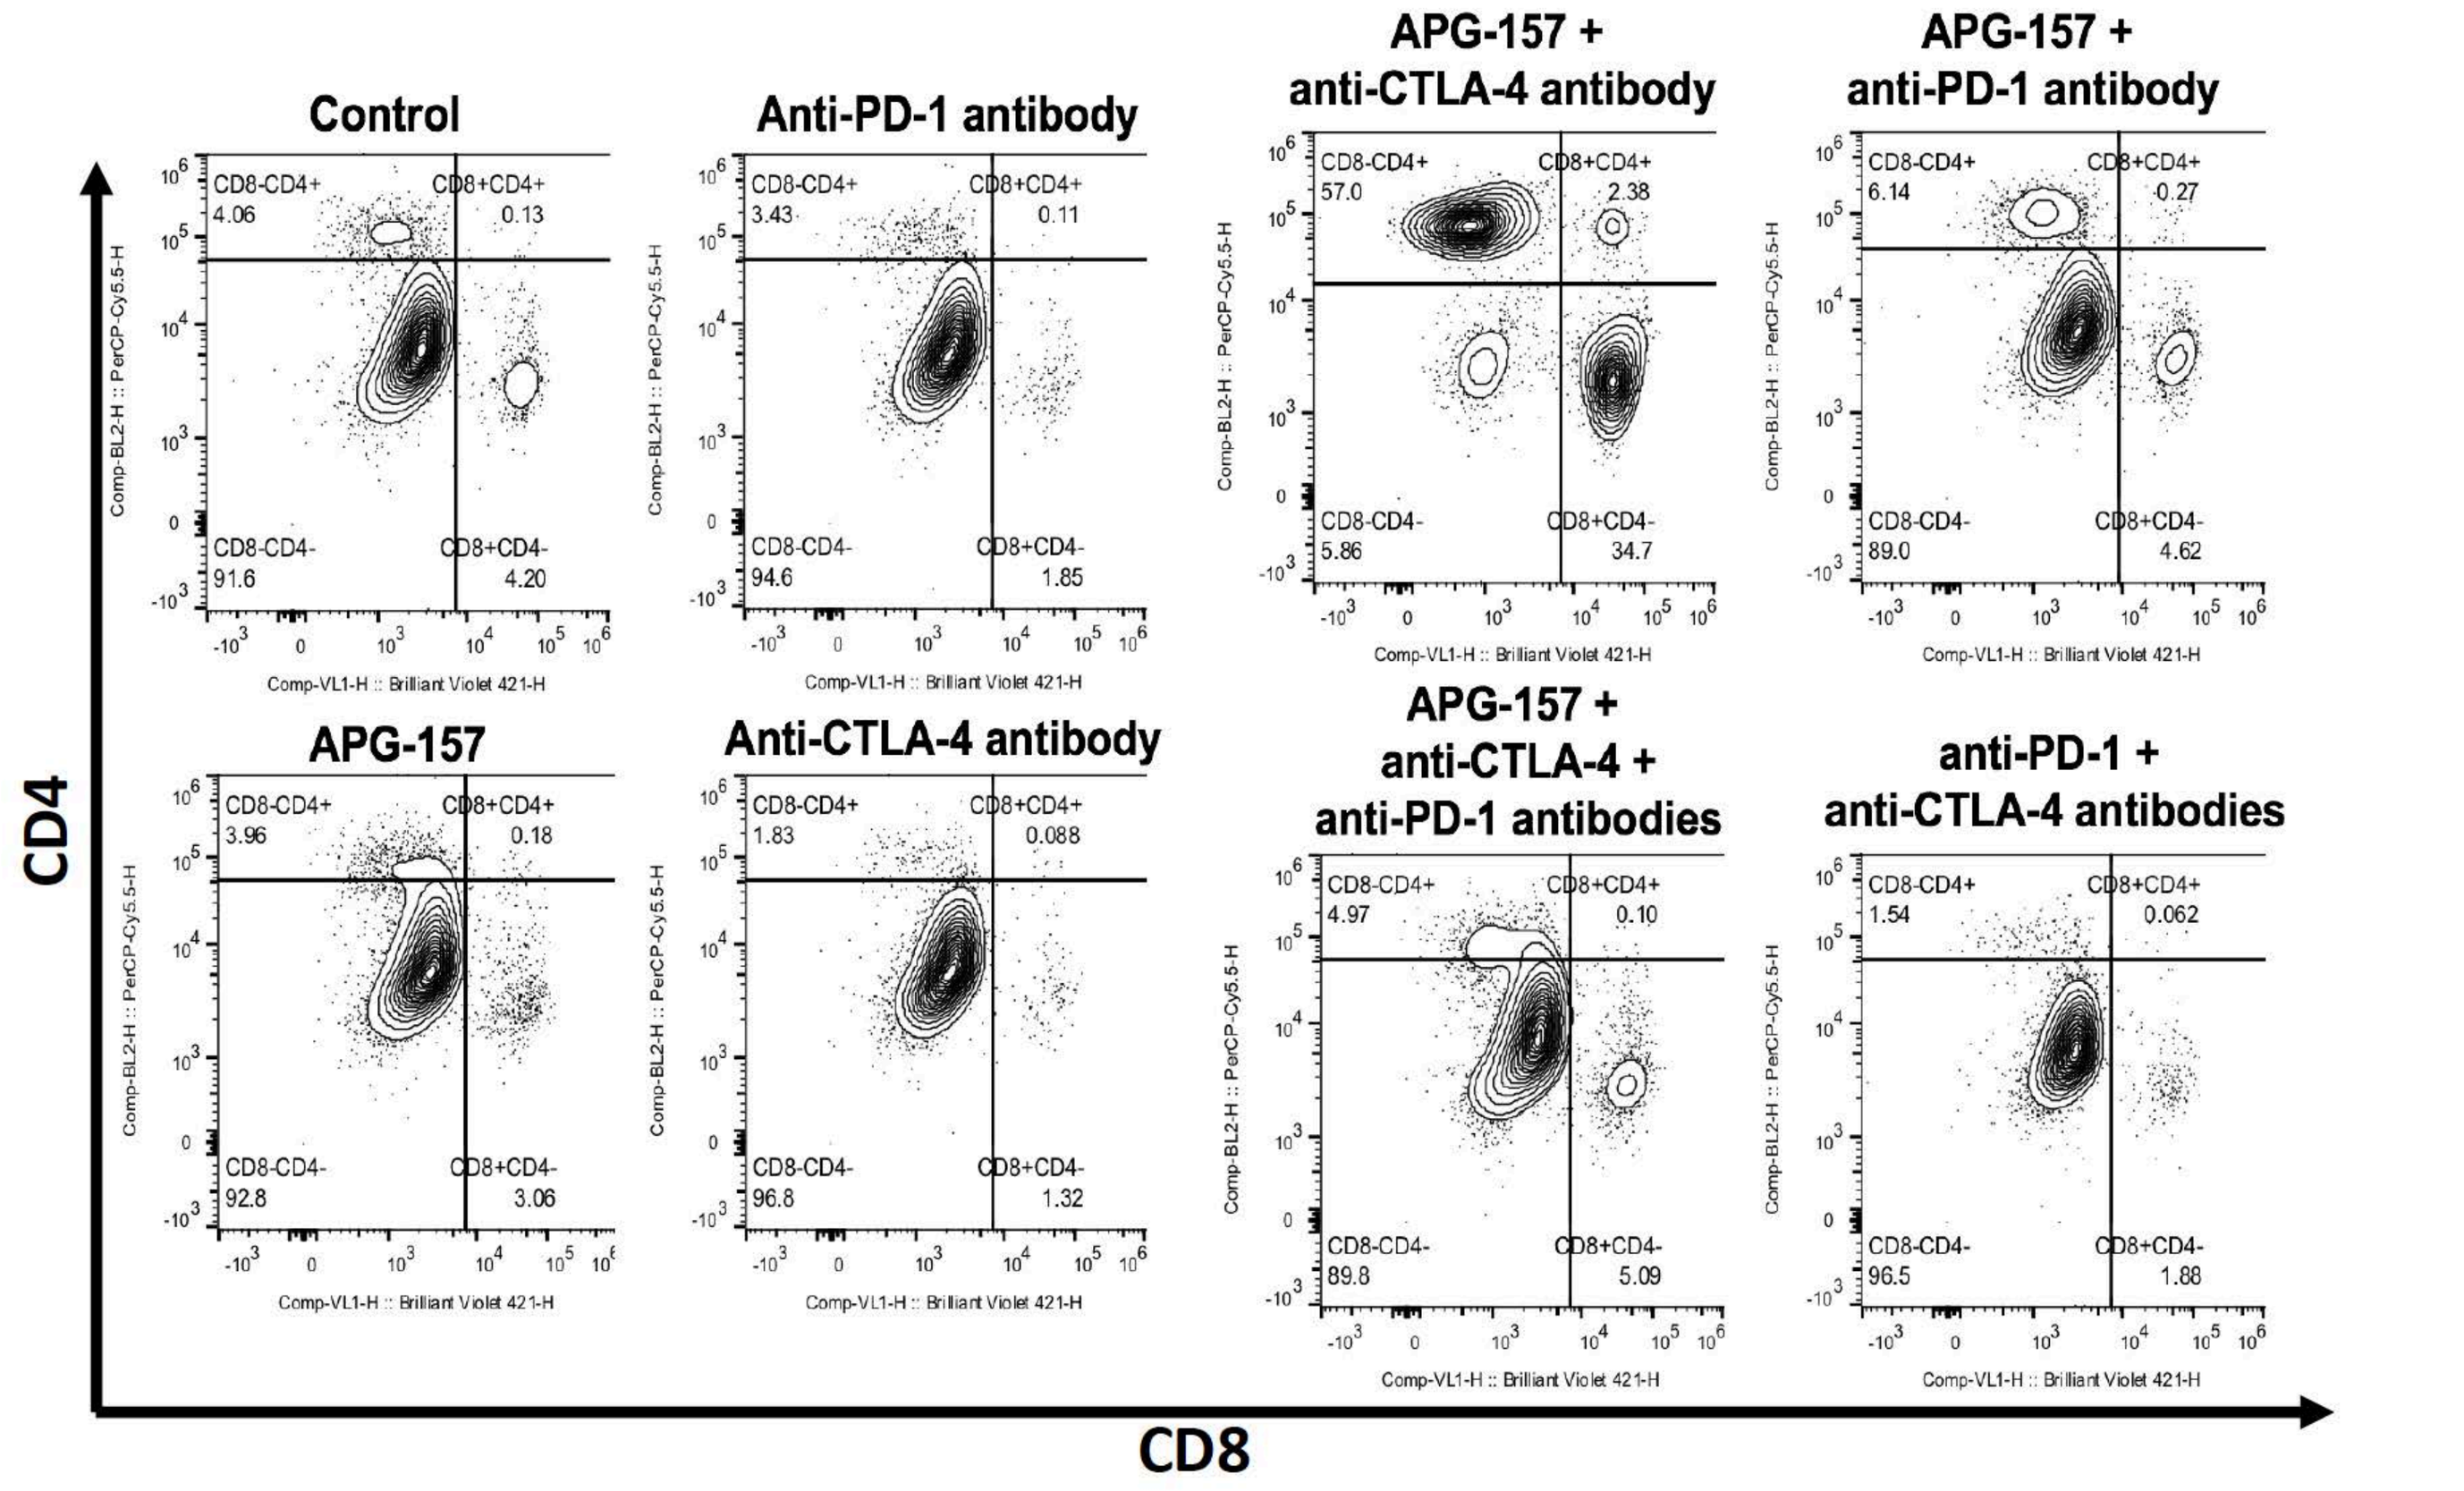
Control
Anti-PD-1 antibody
APG-157 + anti-CTLA-4 antibody
APG-157 + anti-PD-1 antibody
APG-157
Anti-CTLA-4 antibody
APG-157 + anti-CTLA-4 + anti-PD-1 antibodies
anti-PD-1 + anti-CTLA-4 antibodies
CD4
CD8

- The immunophenotype was assessed by multi-color flowcytometry and APG-157 combined with anti-CTLA-4 antibody group showed highest percentage of CD8+ T cells (gated by live cells, subsequent CD3+ T cell population. Not only CD8+ T cells, there was substantial increase in CD4+ T cell population with this group.

Figure 5: RNA-seq of tumor revealed significant down-regulation of EMT pathway genes with APG-157 and anti-CTLA-4 antibody treatment.

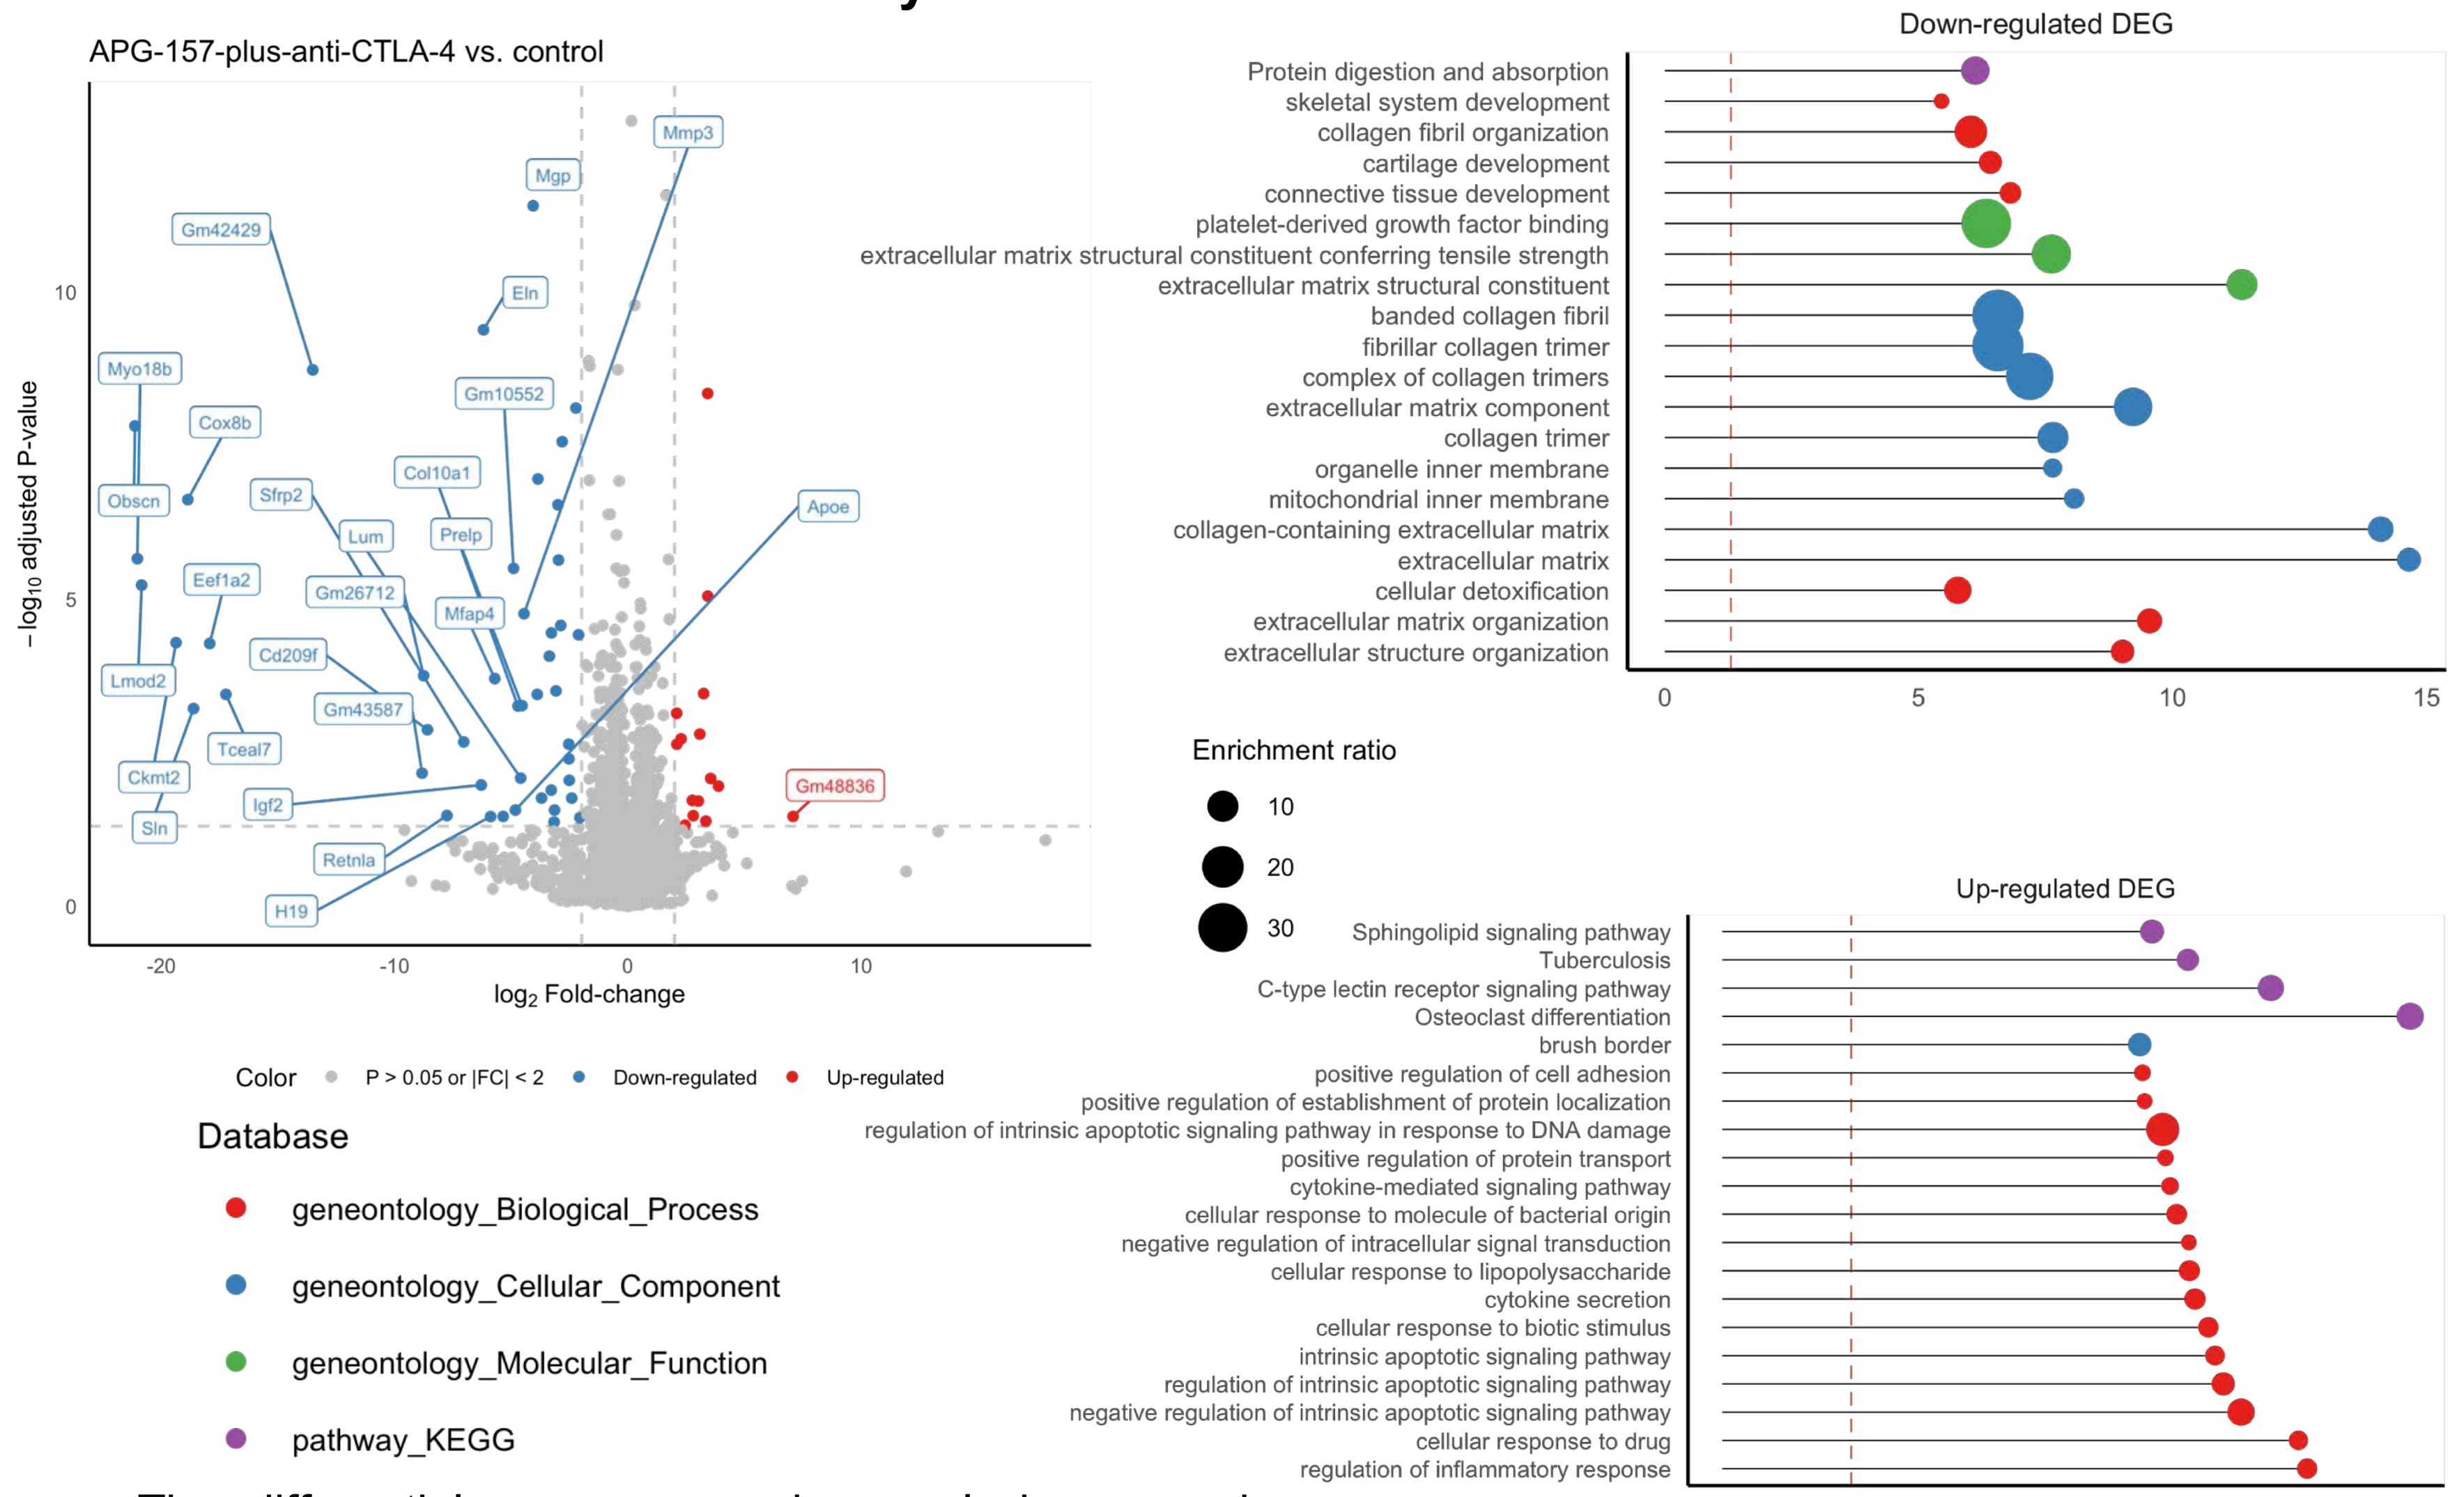
APG-157-plus-anti-CTLA-4 vs. control
log₂ Fold-change
−log₁₀ adjusted P-value
Color P > 0.05 or |FC| < 2 Down-regulated Up-regulated
Down-regulated DEG
Up-regulated DEG
Enrichment ratio
Database
geneontology_Biological_Process
geneontology_Cellular_Component
geneontology_Molecular_Function
pathway_KEGG
−log₁₀ P-value

- The differential gene expression analysis comparing control vs APG-157 and anti-CTLA-4 treatment groups revealed significant down-regulation of EMT/extracellular matrix remodeling related genes and up-regulation of immune responsive genes which might explain the improved tumor control among other groups.

Figure 6: CD8+ T cell depletion abolishes anti-tumor activity of APG-157 and anti-CTLA-4 treatment.

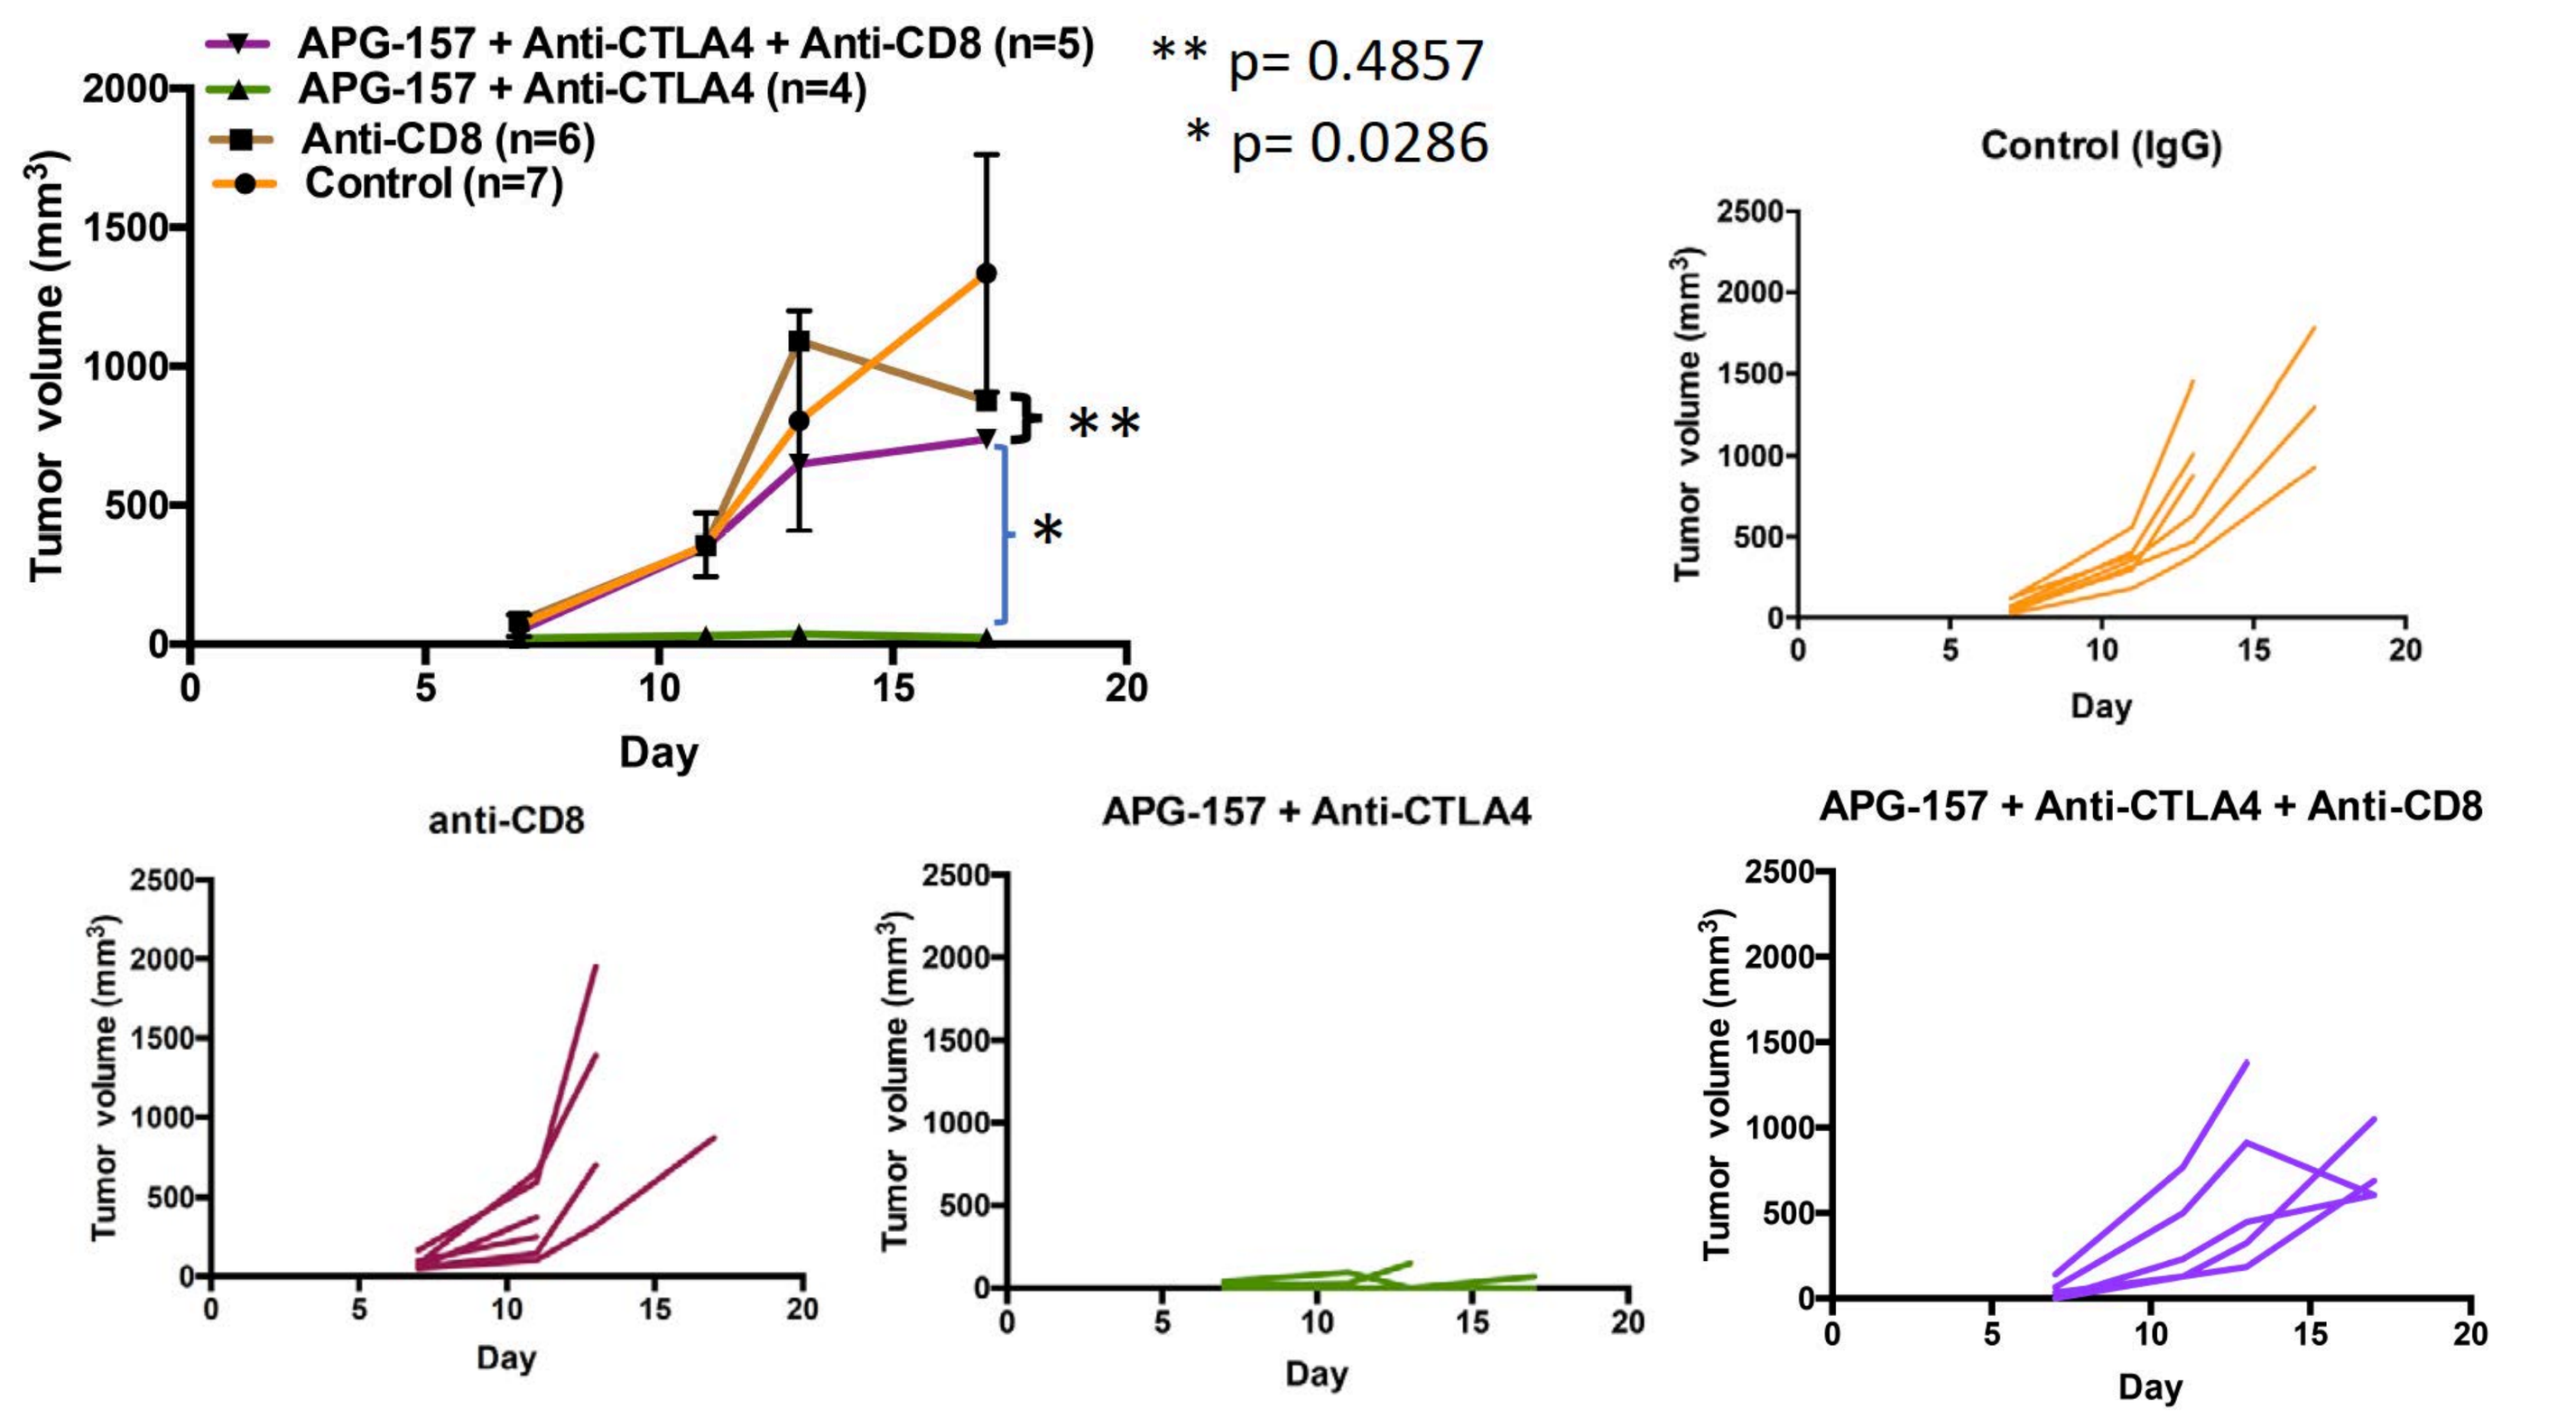
APG-157 + Anti-CTLA4 + Anti-CD8 (n=5)
APG-157 + Anti-CTLA4 (n=4)
Anti-CD8 (n=6)
Control (n=7)
** p= 0.4857
* p= 0.0286
Tumor volume (mm³)
Day
Control (IgG)
anti-CD8
APG-157 + Anti-CTLA4
APG-157 + Anti-CTLA4 + Anti-CD8

Figure 7: Gut microbial changes mediated by APG-157 and immune checkpoint inhibitors

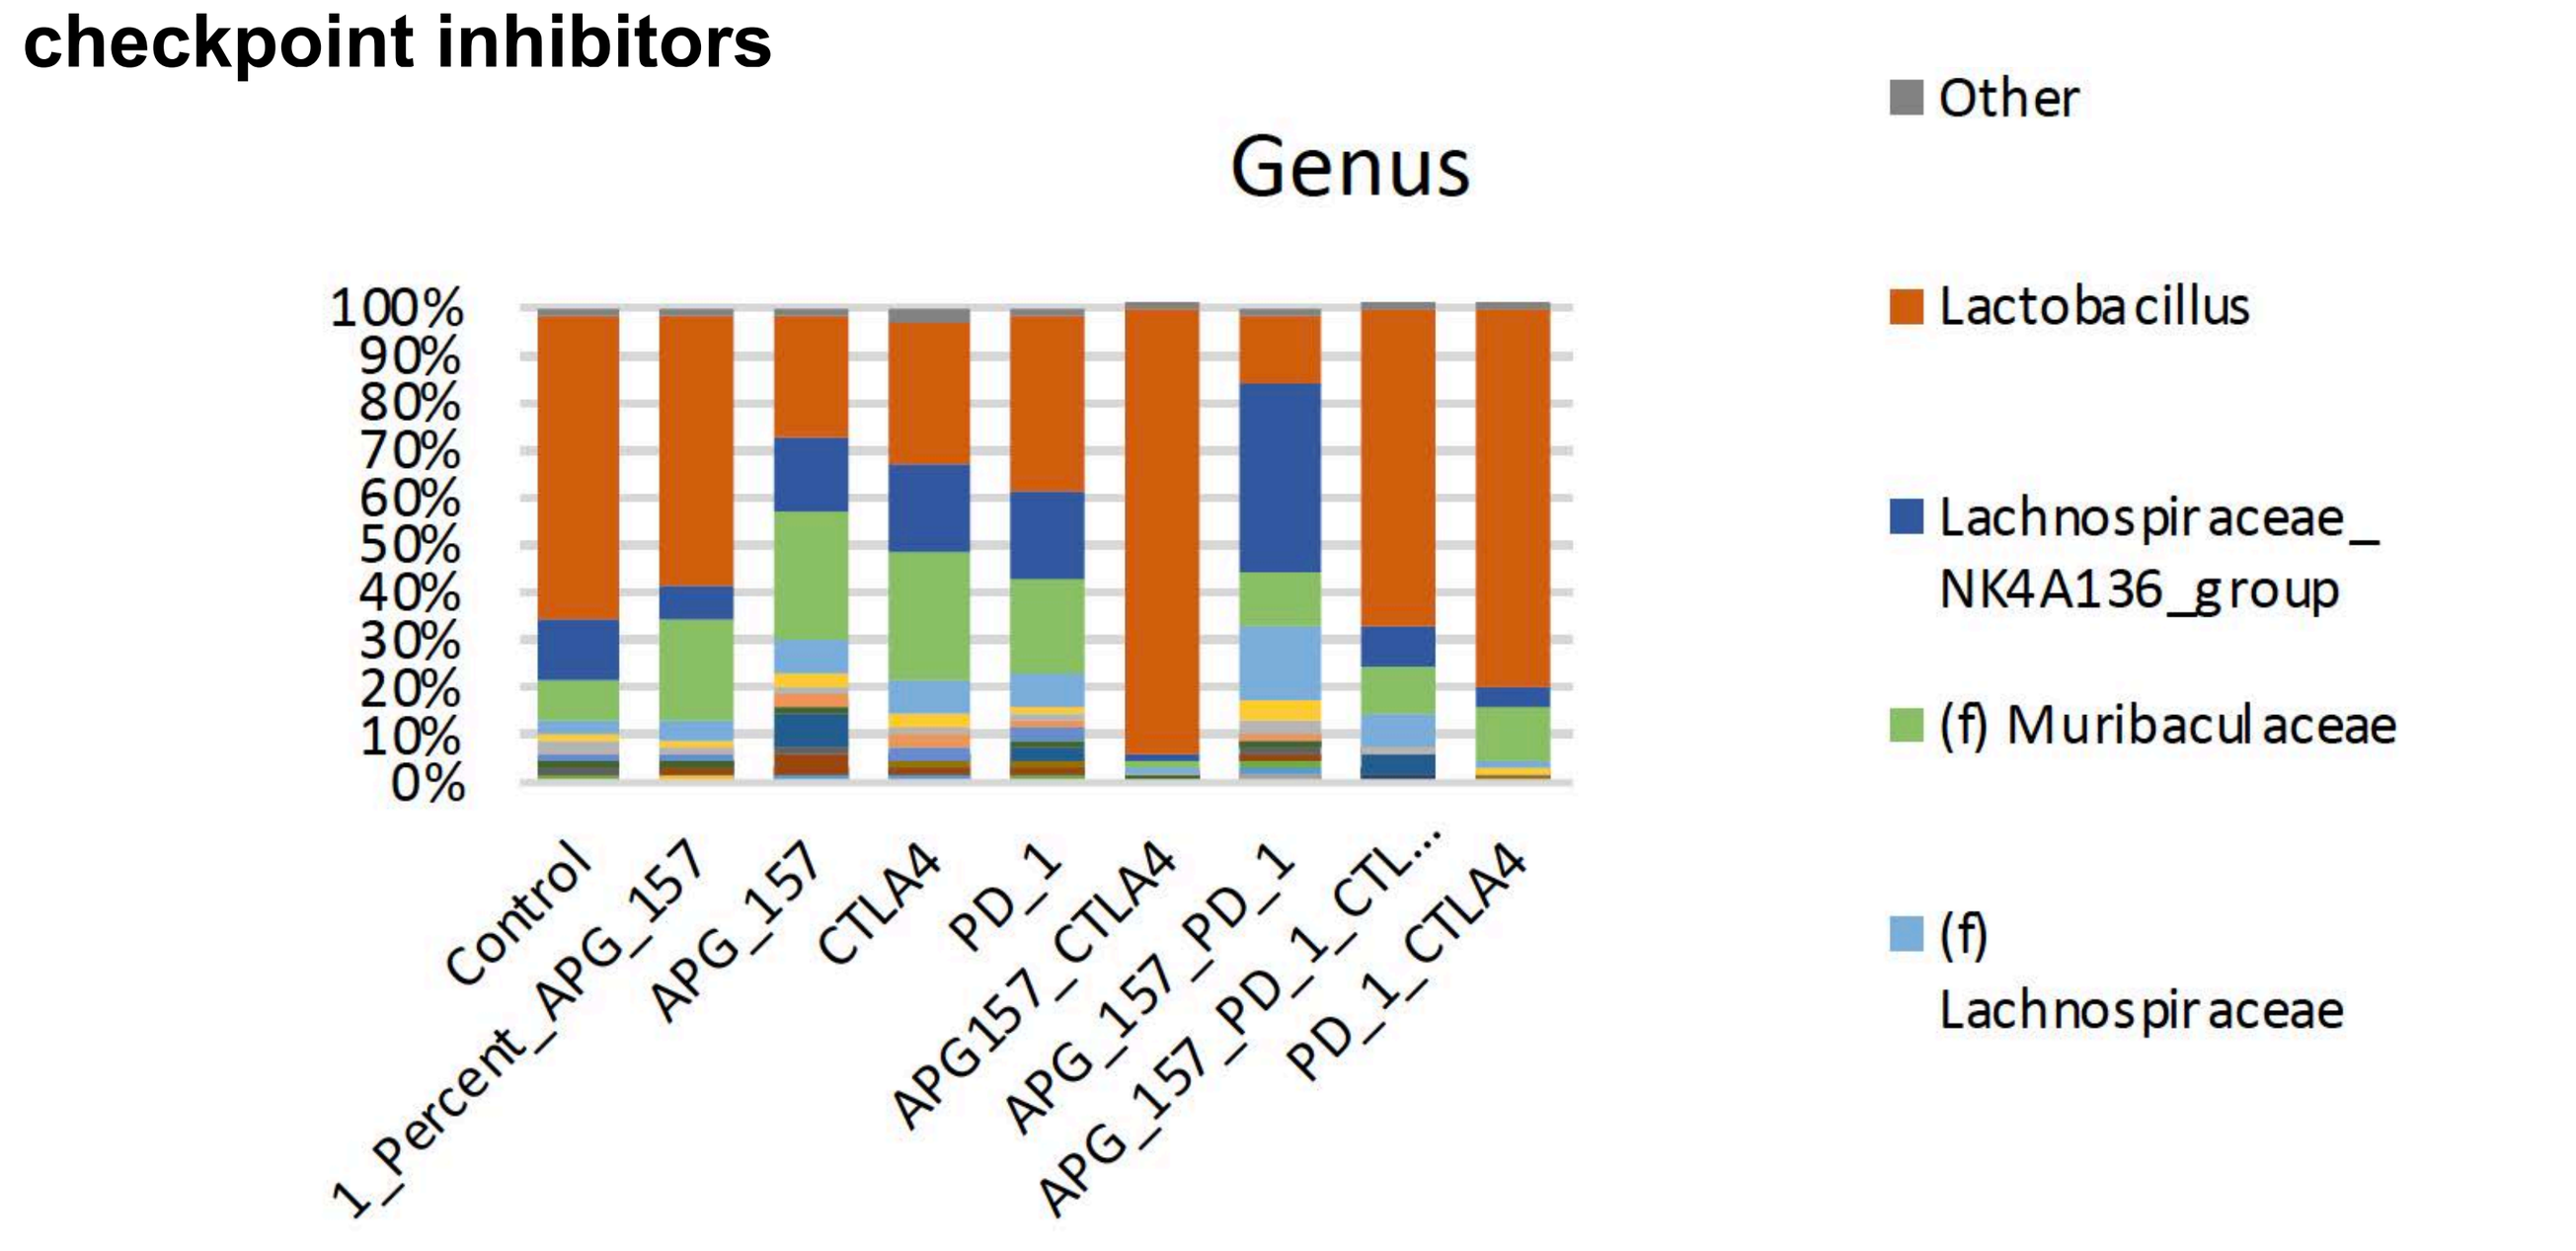
Genus
Other
Lactobacillus
Lachnospiraceae_NK4A136_group
(f) Muribaculaceae
(f) Lachnospiraceae
Control
1_Percent_APG_157
APG_157
CTLA4
PD_1
APG157_CTLA4
APG_157_PD_1
APG_157_PD_1_CTL...
PD_1_CTLA4

- Gut microbiome was assessed by 16sRNA sequence from the feces we collected for each treatment group. There was substantial increased in Lactobacillus for APG-157 and anti-CTLA-4 antibody treatment group.

CONCLUSIONS

- The data supports immunomodulatory effect of APG-157 and anti-CTLA-4 antibody treatment that result in increased immune cell infiltration.
- Gut microbiome change mediated by APG-157 and anti-CTLA-4 warrants further investigation to understand mechanism and contribution to anti-tumor activity.
- This study supports further clinical investigation using APG-157 and immune checkpoint inhibitor combination treatment for head and neck cancer .

REFERENCES

Basak SK, Bera A, Yoon AJ, Morselli M, Jeong C, Tosevska A, Dong TS, Eklund M, Russ E, Nasser H, Lagishetty V, Guo R, Sajed D, Mudgal S, Mehta P, Avila L, Srivastava M, Faull K, Jacobs J, Pellegrini M, Shin DS, Srivatsan ES, Wang MB. A randomized, phase 1, placebo-controlled trial of APG-157 in oral cancer demonstrates systemic absorption and an inhibitory effect on cytokines and tumor-associated microbes. Cancer. 2020 Feb 5. doi: 10.1002/cncr.32644.

Kim, S.G., Veena, M.S., Basak, S. K., Han, E., Tajima, T., Gjertson, D.W., Starr, J., Eidelman, O., Pollard, H.B., Srivastava, M., Srivatsan, E.S., and Wang, M.B. Curcumin Treatment Suppresses IKKβ Kinase Activity of Salivary Cells of Patients with Head and Neck Cancer: A Pilot Study. Clinical Cancer Res. 17:5953-5961, 2011. PMCID: PMC3176971.

Duarte, V.M., Han, E., Veena, M.S., Salvado, A., Suh, J.D., Liang, L-J, Faull, K.F., Srivatsan, E.S., and Wang, M.B. Curcumin enhances the effect of cisplatin in the suppression of head and neck squamous cell carcinoma via the inhibition of IKKβ protein of the nuclear factor kB (NFκB) pathway. Mol. Cancer Therapeutics 9:2665-2675, 2010. PMCID: PMC3004013