6087

Neoadjuvant APG-157 monotherapy in patients with locally advanced squamous cell carcinoma of head and neck: A phase IIA, Single Arm Trial

Marilene B. Wang, MD[1,2], Saroj K. Basak, PhD[2], Eri S. Srivatsan, PhD[2,3], Daniel Sanghoon Shin, MD[4], Saman Hazany, MD[2,3,10], Matteo Pellegrini, PhD[5], Jin Zhong, MD[6], Georgia Del Vecchio, PhD[5], Jonathan Perrie[6], Neda A. Moatamed[8], Luis Z. Avila, PhD[9], Parag Mehta, PhD[9], Selda Samakoglu, MD, PhD[9], Mirian Markley[11,12], Elizabeth Franzmann, MD[11,12]

[1]Department of Head and Neck Surgery, David Geffen School of Medicine at UCLA, CA; [2]Department of Surgery, VA Greater Los Angeles Healthcare System, Los Angeles, CA; [3]Department of Surgery, David Geffen School of Medicine at UCLA, CA; [4]Department of Medicine/Hematology-Oncology, David Geffen School of Medicine at UCLA, CA/VA Greater Los Angeles Healthcare System, Los Angeles, CA; [5]Department of Molecular, Cellular and Developmental Biology, UCLA, CA; [6]Department of Pathology and Laboratory Medicine, VA Greater Los Angeles Healthcare System, Los Angeles, CA; [7]Bioinformatics Interdepartmental Program, UCLA, Los Angeles, CA; [8]Department of Pathology and Laboratory Medicine, David Geffen School of Medicine at UCLA, CA; [9]Aveta Biomics, Inc., 110 Great Rd, Ste 302, Bedford, MA 01730; [10]University of Southern California, Keck School of Medicine, Los Angeles, CA; [11]Department of Otolaryngology, Miller School of Medicine, University of Miami, Miami, Florida; [12]Sylvester Comprehensive Cancer Center, Miller School of Medicine, University of Miami, Miami, FloridaCenter, Miller School of Medicine, University of Miami, Miami, Florida

BACKGROUND

Head and Neck Squamous Cell Carcinoma (HNSCC) carries significant morbidity and mortality. In 2024, over 71,000 new U.S. cases and 16,000 deaths are expected. Most patients present with **locally advanced HNSCC (LA-HNSCC),** where five-year survival rates remain approximately 40-50% in oral cavity and 30-40% (HPV-) or 60-70% (HPV+) oropharyngeal cancer despite aggressive treatment.

A 4–6-week treatment delay can cause tumor growth and upstaging due to HNSCC's rapid progression. Despite curative surgery and chemoradiotherapy, 25–50% of patients—especially HPV-negative—recur within a year. Current treatments are highly morbid, often leading to disfigurement, speech loss, and swallowing difficulties.

No FDA-approved neoadjuvant therapies exist for LA-HNSCC. An effective, safe neoadjuvant option in newly diagnosed patients could:

- Shrink or stabilize tumors to enable de-intensified surgery, radiation, or chemotherapy
- Preserve speech/swallowing and reduce treatment-related morbidity
- Lower recurrence risk and improve event-free & long-term survival

PHASE 2A STUDY (NCT05312710) DESIGN

Diagnosis of cancer — APG-157 Administration for 4 to 6 Weeks — **Definitive treatment**

Orally given 600 mgs (200 mgsX3 times) / day, supported by PK data

STUDY OBJECTIVES

Stop tumor growth and/or shrink tumor
Modulate tumor microenvironment to improve success of definitive treatment
Reduce post-treatment morbidity and increase survival

BASELINE CHARACTERISTICS (n=24)

Sex, n(%)	
Male	20 (83.3%)
Female	4 (16.7%)
Median (min, max) age,	65 (42, 89)

Race, n(%)	
White	15 (62.5%)
Hispanic	2 (8.3%)
African American	6 (25%)
African Indian	1 (4.2%)

Smoker, n (%)	
Current + Cannabis	11 (45.8%) + 2(8.3%)
Former +/- Cannabis	5 (20.8%)
Never	6 (25%)

Primary tumor site, n(%)	
Oral cavity	13 (54.2%)
Oropharynx	11 (45.3%)

P16 status	
Negative	9 (37.5%)
Positive	14 (58.3%)
Equivocal	1 (4.2%)

Clinical T staging	
T1	5 (20.8%)
T2	7 (29.2%)
T3	7 (29.2%)
T4	5 (20.8%)

Clinical N staging	
N0	9 (37.5%)
N1	11 (45.8%)
N2	4 (16.7%)

Overall staging	
Stage I or II	11 (45.8%)
Stage III or IVA	13 (54.2%)

RESULTS

EVENT FREE SURVIVAL (EFS)

- 96% (23/24) alive & recurrence-free at median 90.1-week follow-up
- One death due to a non-cancer-related cause
- 11 recurrence-free ≥1 year, 10 ≥2 years

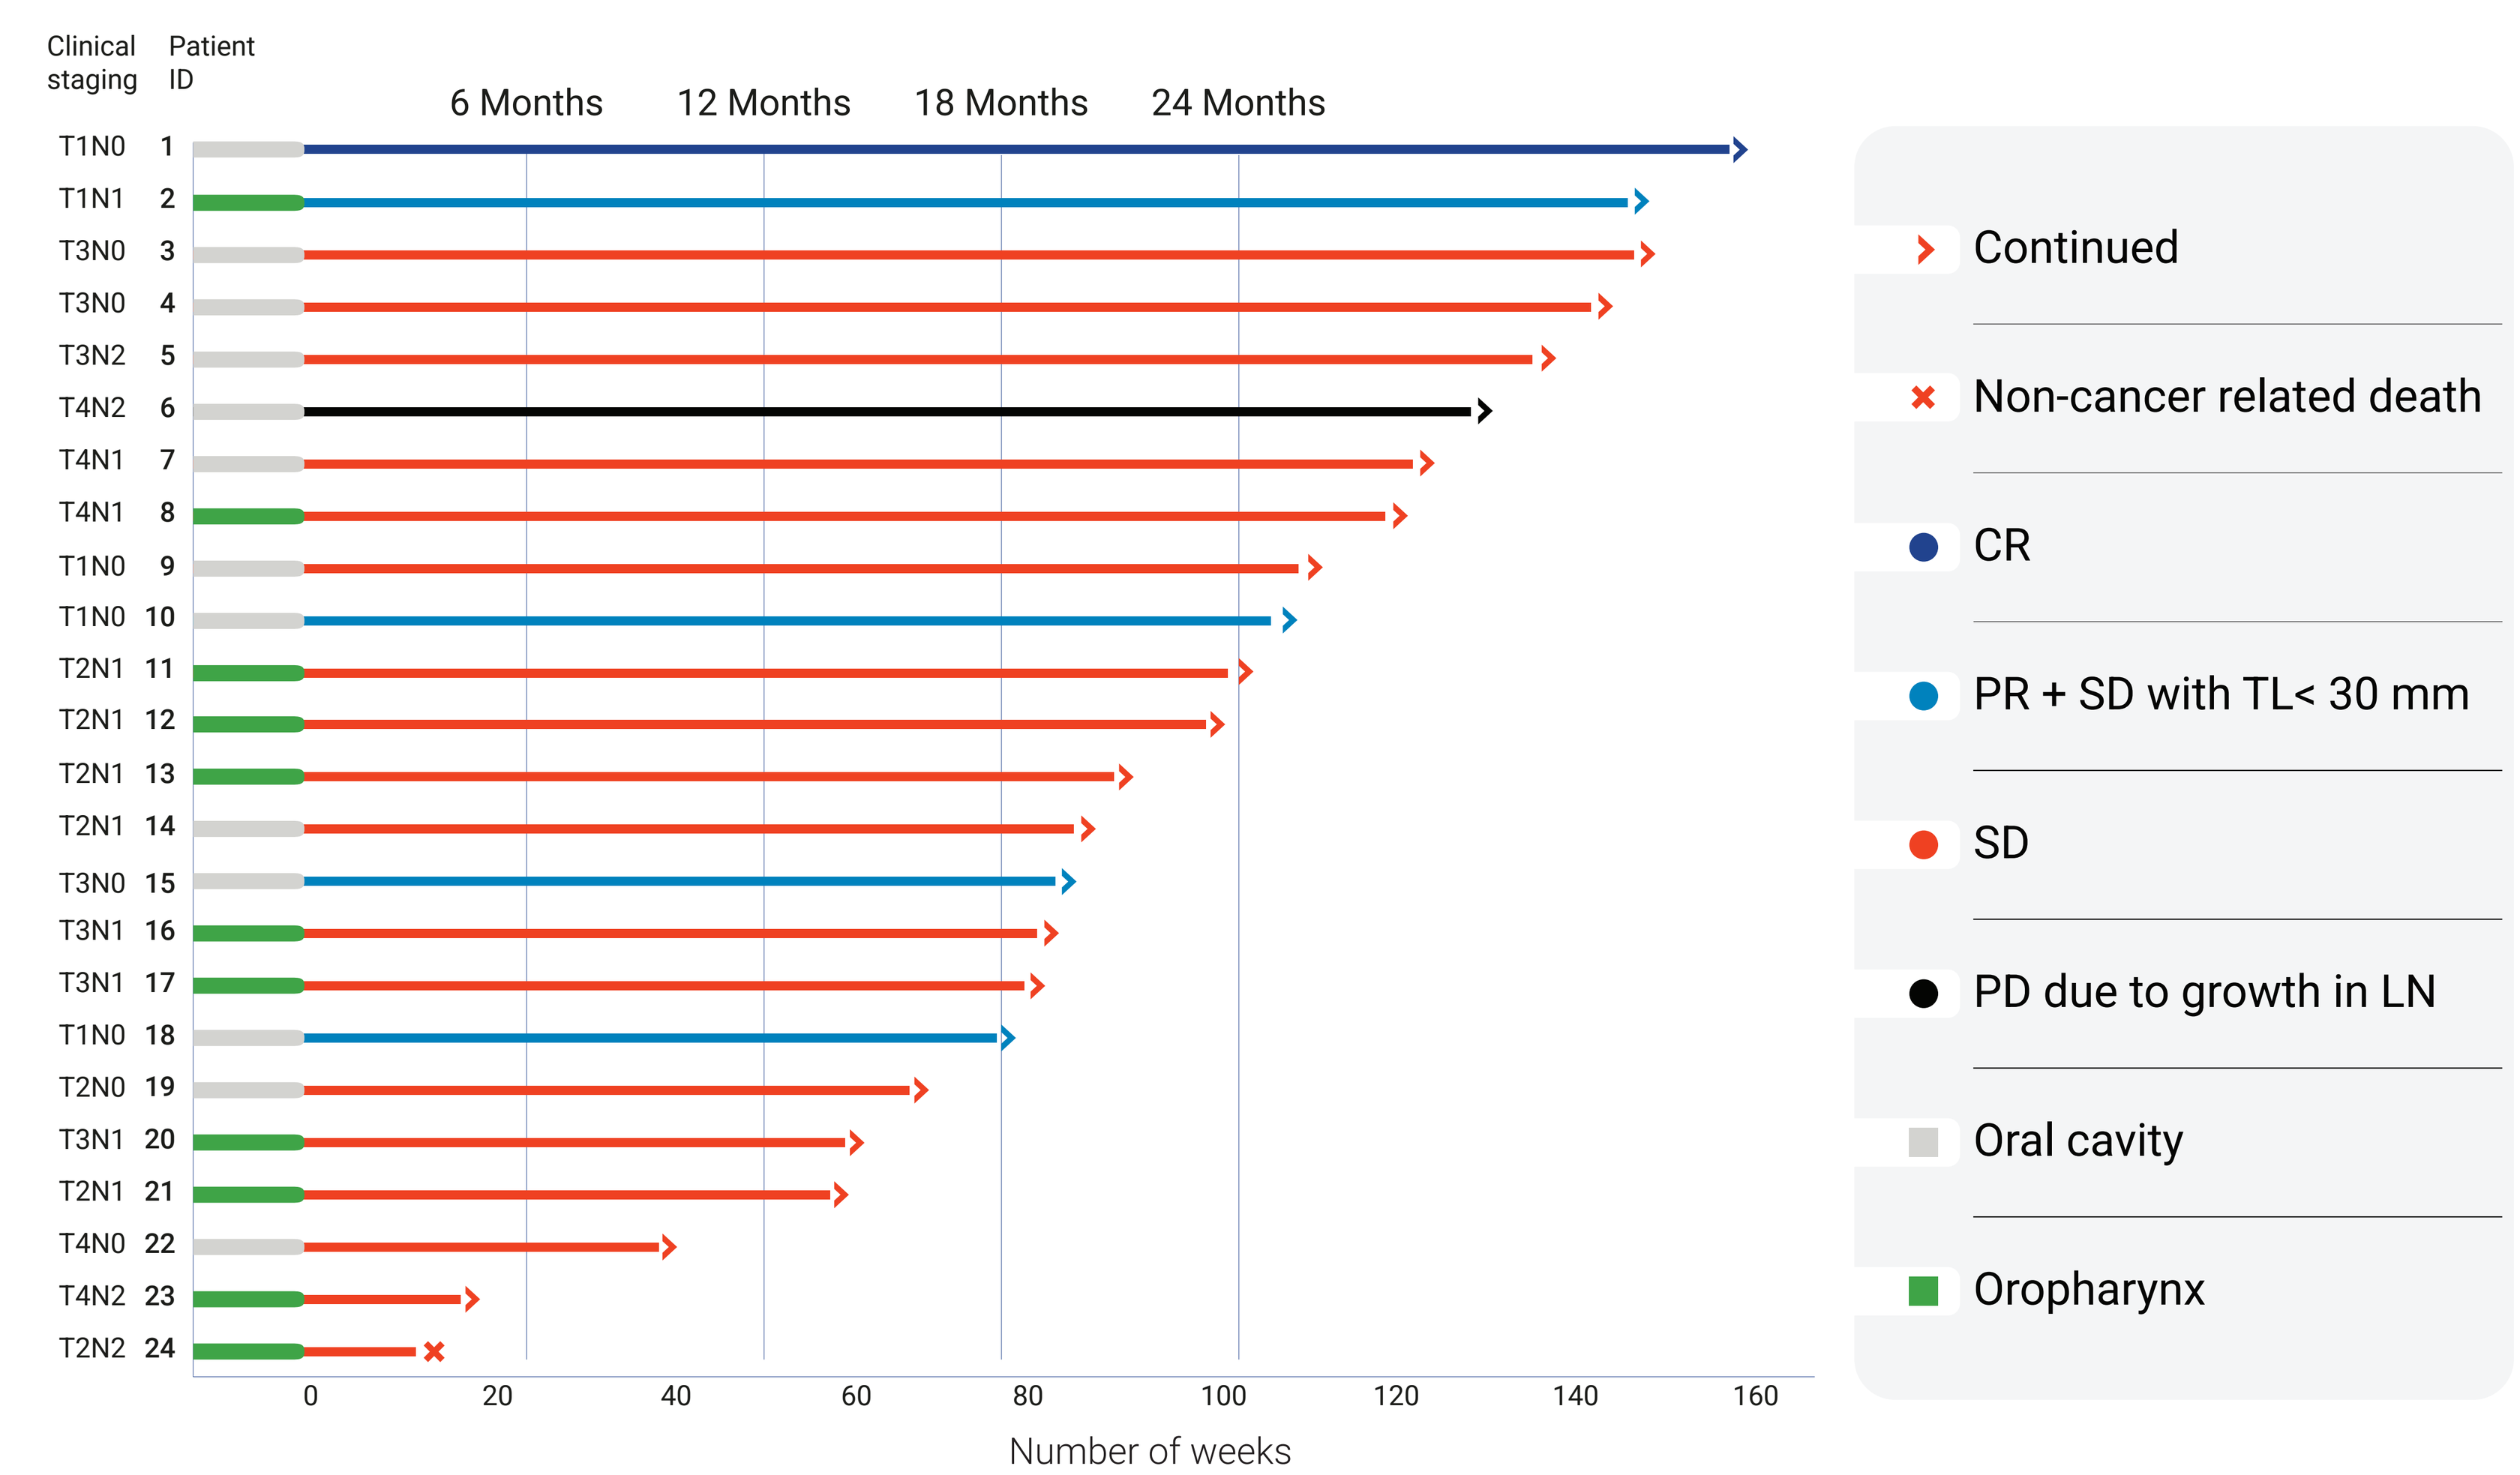

Clinical staging
Patient ID
6 Months
12 Months
18 Months
24 Months
T1N0 1
T1N1 2
T3N0 3
T3N0 4
T3N2 5
T4N2 6
T4N1 7
T4N1 8
T1N0 9
T1N0 10
T2N1 11
T2N1 12
T2N1 13
T2N1 14
T3N0 15
T3N1 16
T3N1 17
T1N0 18
T2N0 19
T3N1 20
T2N1 21
T4N0 22
T4N2 23
T2N2 24
0
20
40
60
80
100
120
140
160
Number of weeks
Continued
Non-cancer related death
CR
PR + SD with TL< 30 mm
SD
PD due to growth in LN
Oral cavity
Oropharynx

ctDNA CLEARANCE ON NEOADJUVANT APG-157

- 80% patients achieved >90% reduction in ctDNA

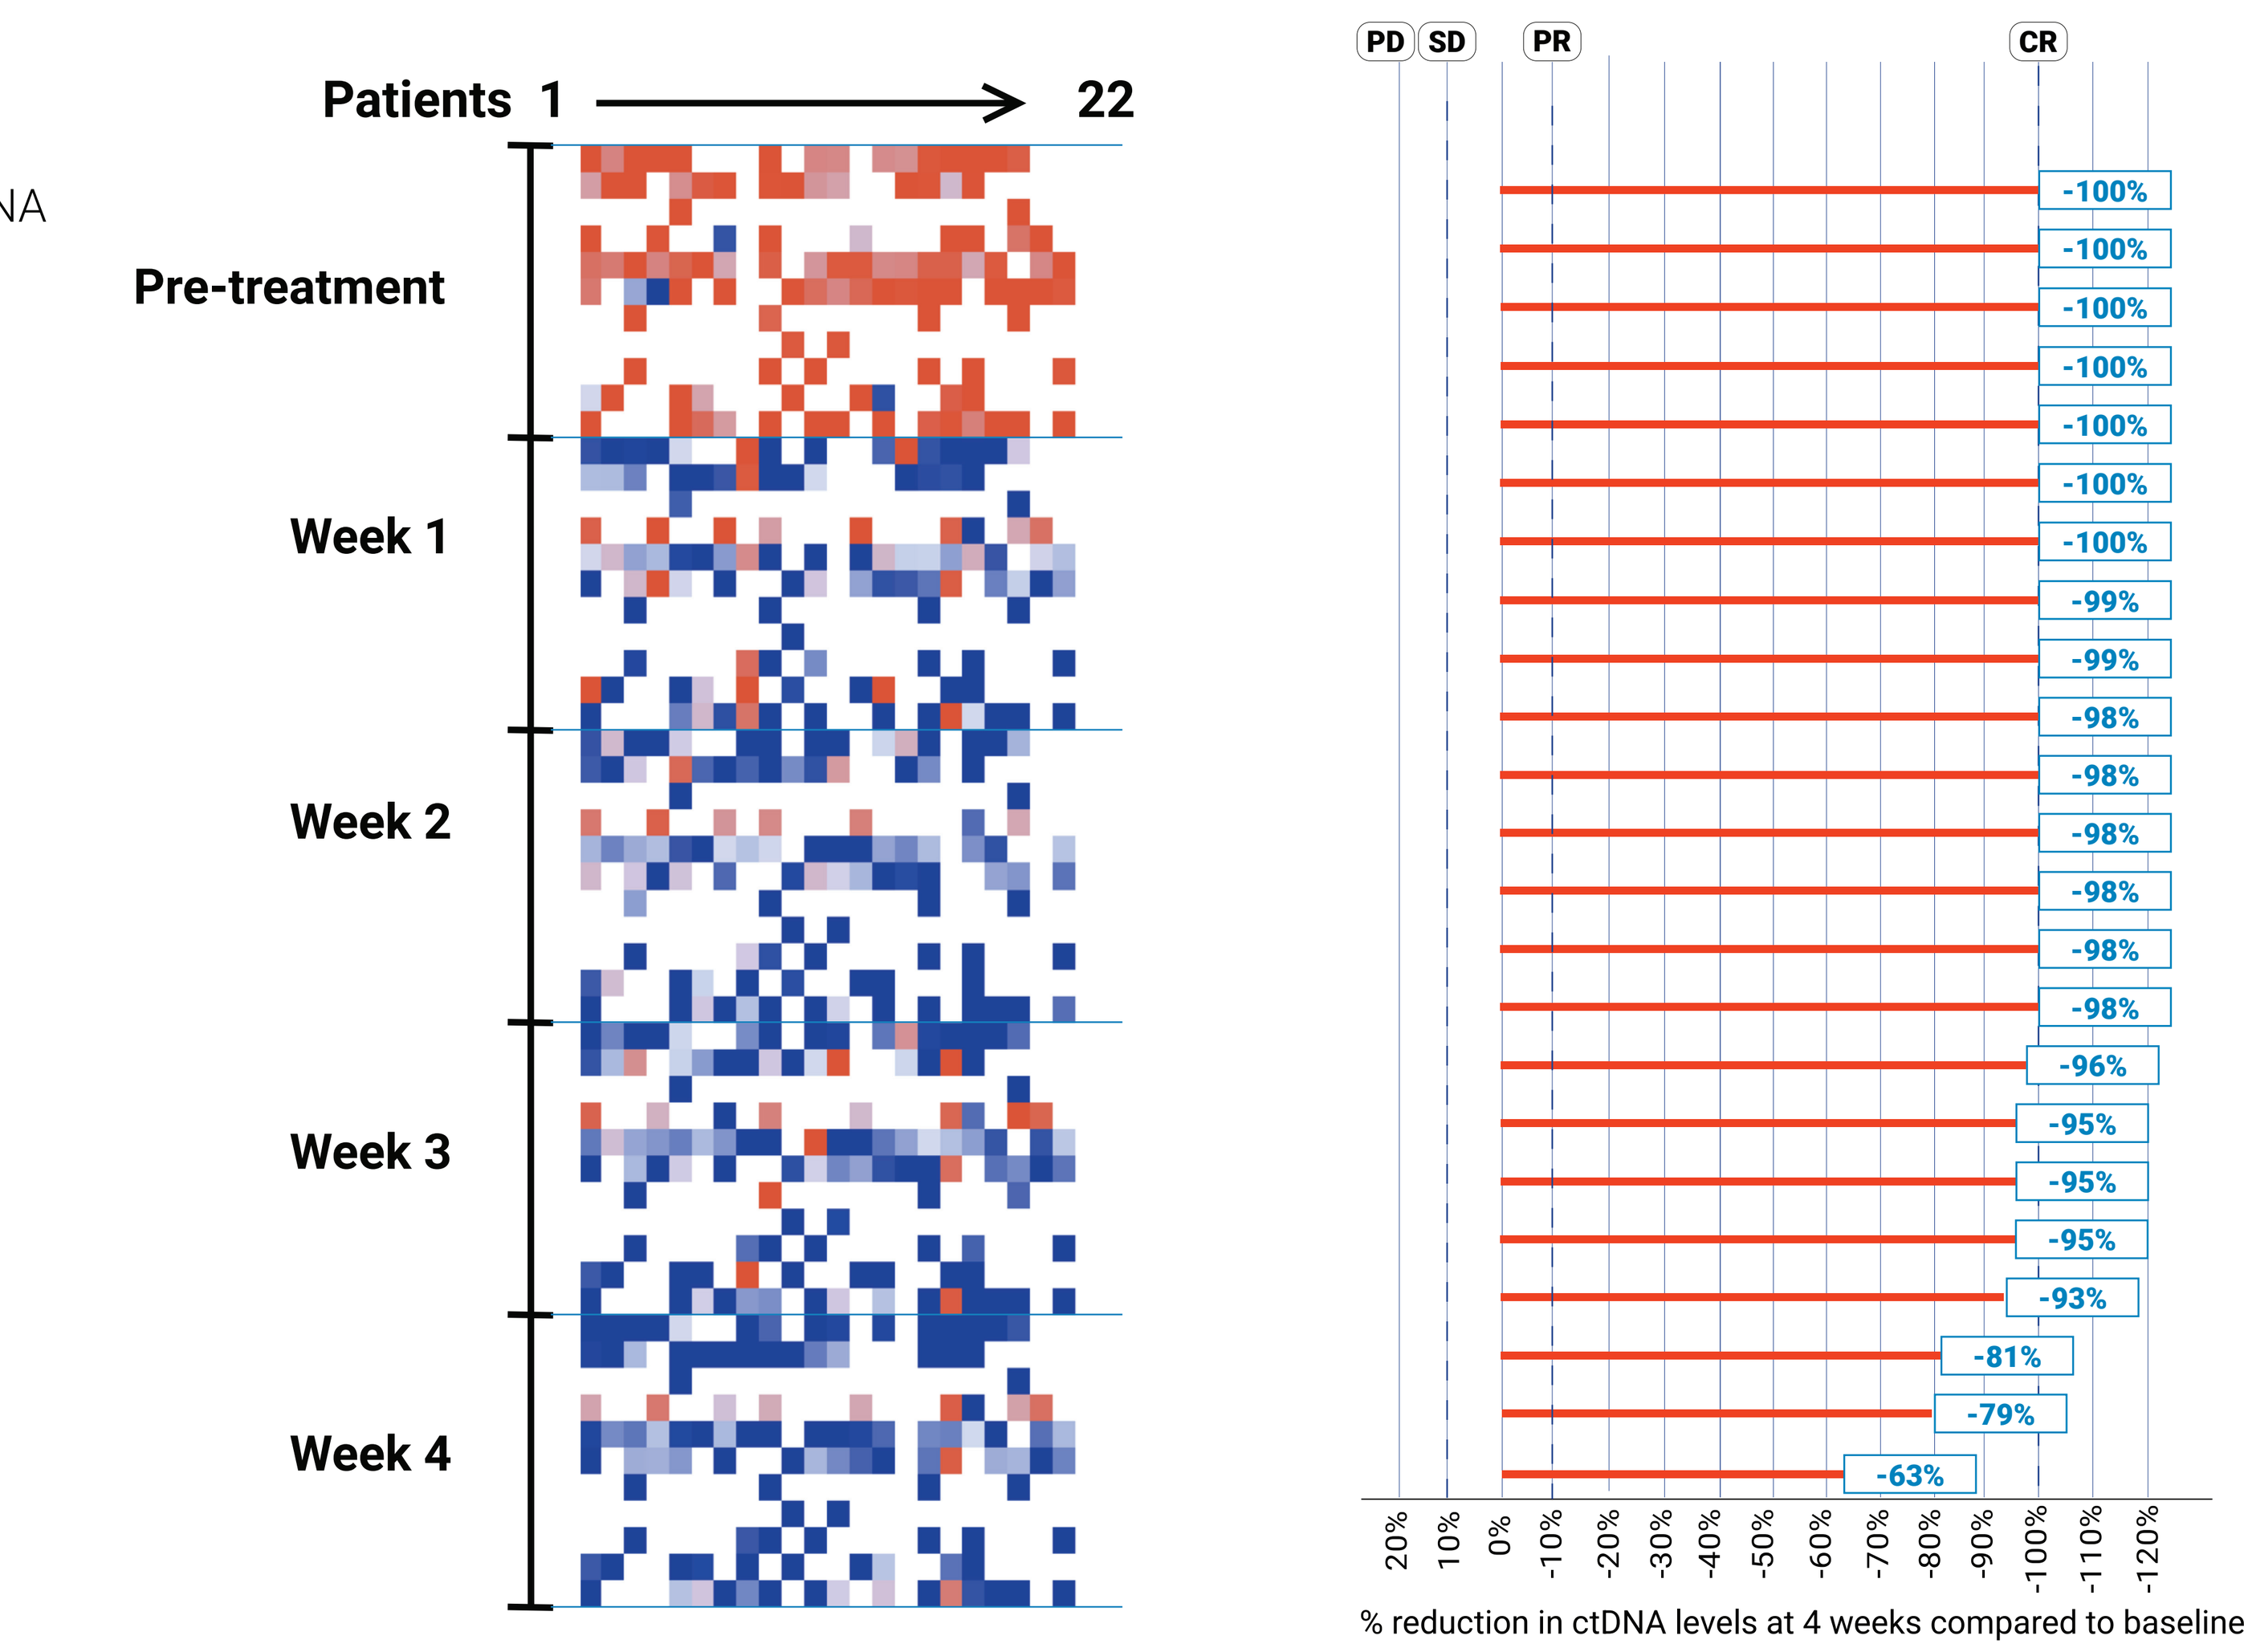

Patients 1 22
Pre-treatment
Week 1
Week 2
Week 3
Week 4
PD
SD
PR
CR
-100%
-100%
-100%
-100%
-100%
-100%
-100%
-99%
-99%
-98%
-98%
-98%
-98%
-98%
-98%
-96%
-95%
-95%
-95%
-93%
-81%
-79%
-63%
20%
10%
0%
-10%
-20%
-30%
-40%
-50%
-60%
-70%
-80%
-90%
-100%
-110%
-120%
% reduction in ctDNA levels at 4 weeks compared to baseline

RESPONSE ASSESSMENT USING RECIST V1.1

- Disease Control Rate (DCR): 95.8%
- Overall Response Rate (ORR): 16.7% with 2 Complete Response (CR) and 2 Partial Response (PR)
- Tumor regression: 45% showed reduction in the size of primary lesion

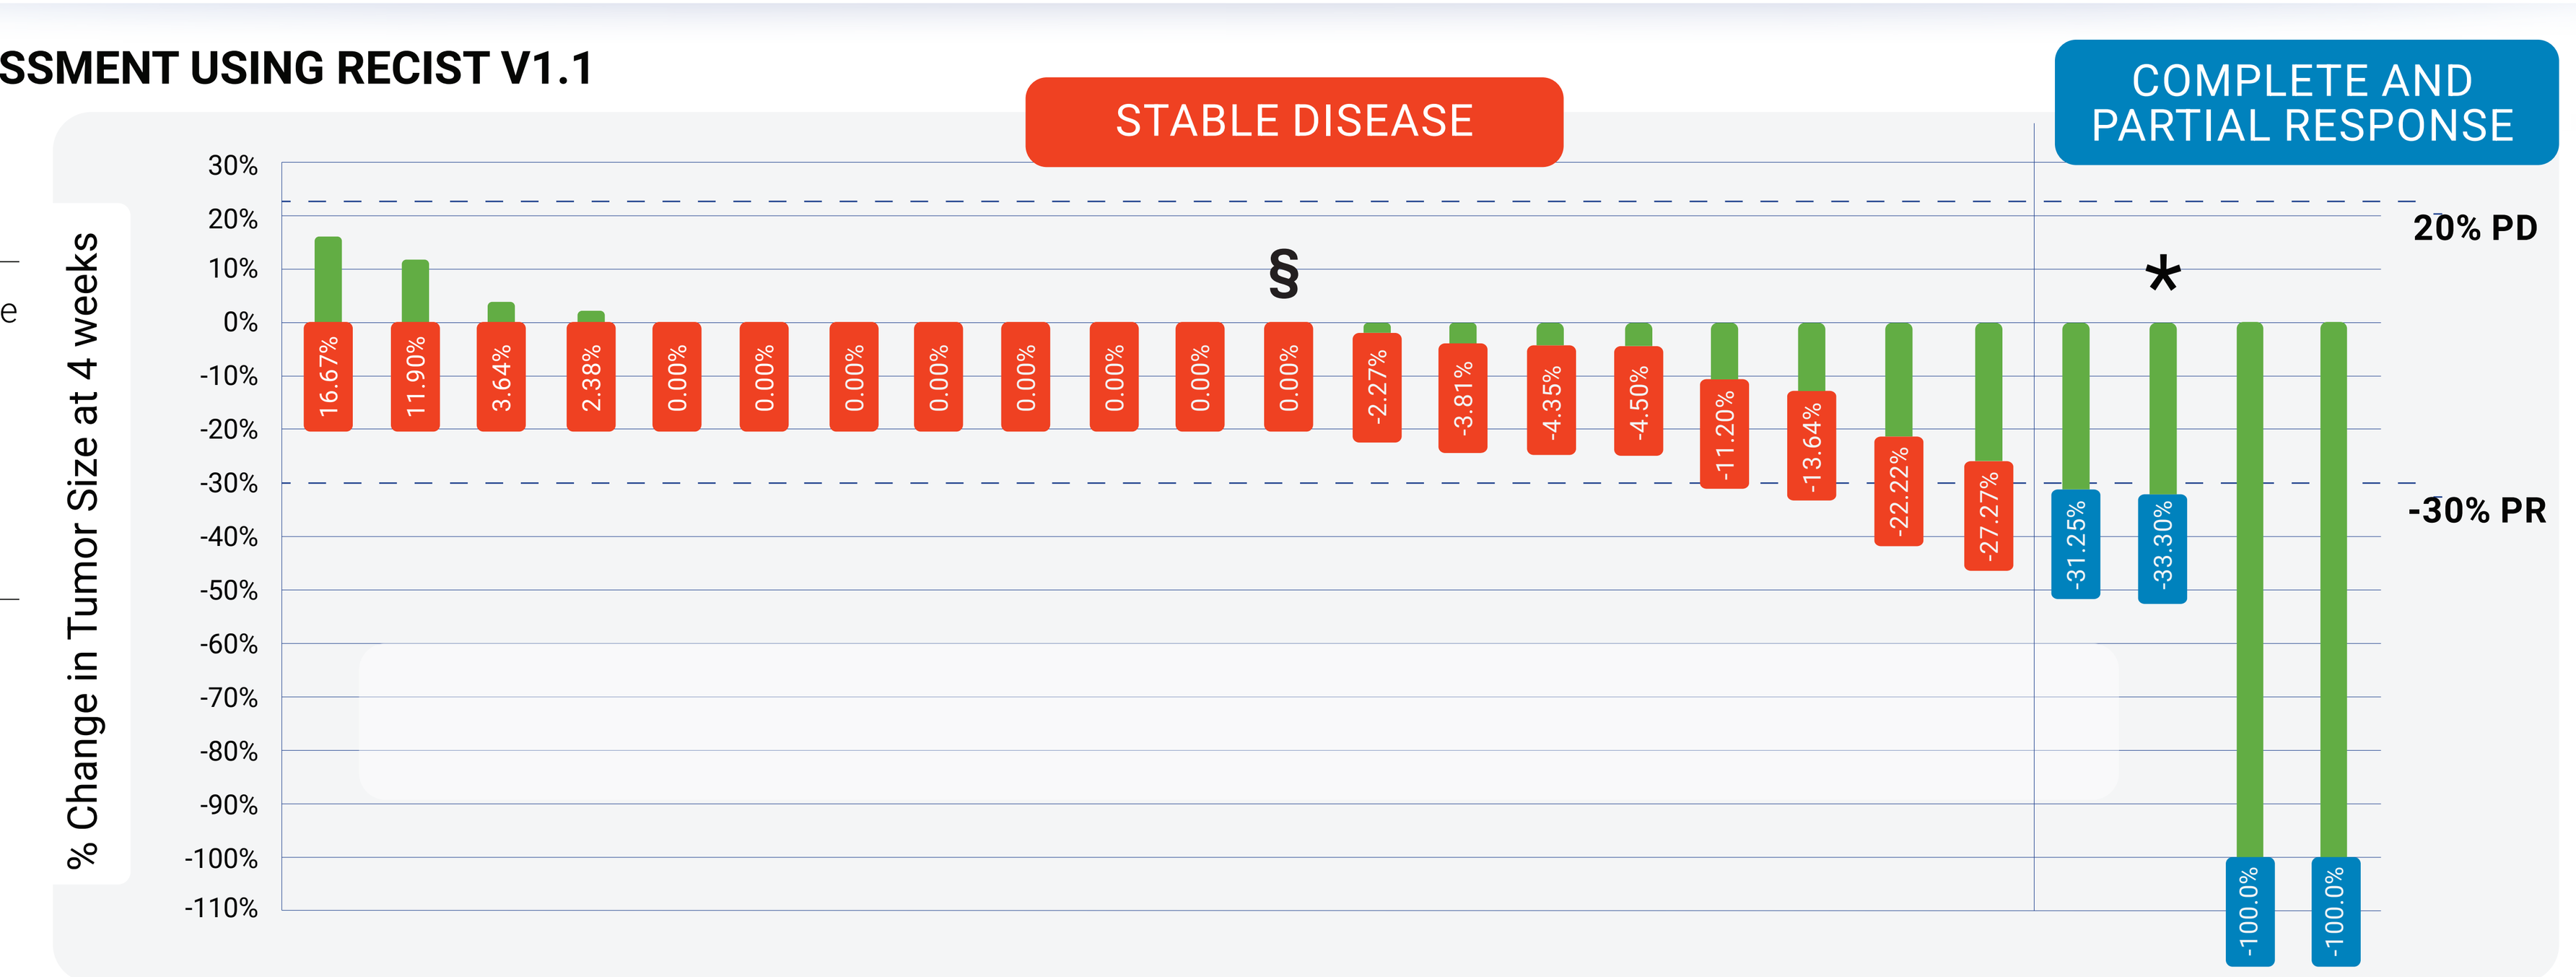

STABLE DISEASE
COMPLETE AND PARTIAL RESPONSE
% Change in Tumor Size at 4 weeks
30%
20%
10%
0%
-10%
-20%
-30%
-40%
-50%
-60%
-70%
-80%
-90%
-100%
-110%
20% PD
-30% PR
16.67%
11.90%
3.64%
2.38%
0.00%
0.00%
0.00%
0.00%
0.00%
0.00%
0.00%
0.00%
-2.27%
-3.81%
-4.35%
-4.50%
-11.20%
-13.64%
-22.22%
-27.27%
-31.25%
-33.30%
-100.0%
-100.0%
§
*

§Target lesion SD, time point response PD due to appearance of a new lymph node lesion; patient previously treated ovarian cancer
*Anti-tumor activity based on pathology as post-treatment radiographic scan was not available.

CLINICAL TO PATHOLOGICAL DOWNSTAGING (Surgical Subsets)

- 46.2% (6 of 13 patients with oral cavity) showed clinical to pathological downstaging

Patient IDs	Pre APG-157 Clinical Staging	Post APG-157 Pathological Staging
1	T1N0M0	pT1N0M0
3	T3N0M0	pT3N0M0
4	T3N0M0	pT3N0M0
5	T3N2M0	pT2N1M0
6	T4N2M0	pT4pN2M0
7	T4N1M0	pT1N0M0
9	T1N0M0	pT0(Tis)N0M0

Patient IDs	Pre APG-157 Clinical Staging	Post APG-157 Pathological Staging
10	T1N0M0	pT0(Tis)N0M0
14	T2N1M0	pT2N0M0
15	T3N0M0	pT1N0M0
18	T1N0M0	pT1N0M0
19	T2N0M0	pT4bN0M0
22	T4N0M0	pT4aN0M0

Downstaging | Stable | Upstaging

CONCLUSIONS

01 **Event-Free Survival -** 96% (23/24) remain alive and recurrence-free at median 90.1-week follow-up. 1 death due to a non-cancer-related cause. 10 of 20 patients who remained alive surpassed the 12-month survival mark had Stage III/V disease

02 **>80% patients achieved >90% ctDNA clearance prior to definitive therapy** as a surrogate to EFS

03 **No drug-related Grade ≥ 3** treatment emergent adverse events (TEAEs).

04 **No primary tumor progression** in any subject while on APG-157. **45% (11/24)** subjects experienced tumor reduction

05 **No delay in subsequent definitive therapy** with tumor-free resection margins

06 **46.2% (6/13)** of surgical patients had **clinical-to-pathological downstaging**

07 **Single agent activity.** Overall Response Rate (ORR) of 16.7% (4/24): CRs and 2 PRs. ORR is 23% in oral cavity, 33% p16 negative oral cavity cancer and 9.1% in oropharyngeal cancer

REFERENCES

1- Xiao R, Ward C M, Yang K. et al. 2018, Cancer; 1; 124(7): 1400- 1414

2 - Honore N, Marcke Cvan, Galot R at al. 2023, Ann Oncol; 34(12): 1175-1186

ACKNOWLEDGMENT

This study was funded by Aveta Biomics, Inc.

Contact information: efranzman@med.miami.edu

"Presented at the 2025 ASCO Annual Meeting (American Society of Clinical Oncology); May 30 to June 3 , 2025; Chicago. Illinois"

"This poster contains non-enabling illustrative data for conceptual discussion only. Technical details are confidential and can be disclosed under mutual NDA/CDA."

Copyright © 2025, Aveta Biomics, Inc. All rights reserved.